PHASE 0 INTRATUMORAL MICRODEVICE STUDY OF FX-909 IN BLADDER CANCER DEMONSTRATES ON-TARGET ANTI-TUMOR ACTIVITY AND IMMUNE MODULATION, ENHANCED IN COMBINATION WITH ANTI-PD-1

Abstract #CT111

Phuong A. Nguyen[1], Peter M. Szabo[1], Joseph Kotler[2], Simon Chow[2], Bijal Kakrecha[1], Nnamdi Onochie[3], Fanni Santa[2], Michaela Bowden[1], Oliver Jonas[2,4], Matthew Mossanen[2,3], Evisa Gjini[1]

[1]Flare Therapeutics, Cambridge, MA; [2]Mass General Brigham, Boston, MA; [3]Dana-Farber Cancer Institute, Boston, MA; [4]Kibur Medical, Boston, MA

FLARE THERAPEUTICS

INTRODUCTION

- Luminal advanced urothelial carcinoma (UC), defined by high PPARγ expression, represents ~65% of UC cases and is associated with poor clinical outcomes[1,2].
- PPARγ acts as a master regulator of luminal UC, promoting tumor growth and an immune-cold tumor microenvironment.
- PPARγ is important for the maturation and function of various immune system-related cell types, such as monocytes/macrophages, as well as immune-mediated inflammation.
- FX-909, a first-in-class oral PPARγ inhibitor, demonstrated early clinical activity and acceptable safety in Phase 1a (NCT05929235), with correlative biomarker analyses indicating immune activation[3-5].
- Here, we report Phase 0 results (NCT06204614) assessing intratumoral effects of FX-909 alone and with anti-PD-1 using intratumoral microdevice (IMD)[6] implantation and retrieval in primary bladder cancer (**Figure 1**).

METHODOLOGY

- Participants had localized bladder cancer (T2, N0, M0) with tumors containing a ≥1 cm lesion and were planned for cystectomy. Eligibility required clinical stability and sufficient tumor volume.
- Data are shown from two patients (Pt1, Pt2), with IMDs remaining in situ for 48-72 hours (**Figure 2**).
- Two FX-909 doses (D1, 200 mg; D2, 350 mg) were tested. The amount of drug delivered by the microdevice was formulated to match the expected intratumor dose achieved during systemic dosing[7].
- Intratumoral drug penetration was assessed by MALDI-TOF, PPARγ[8] and cleaved caspase-3 (CC3) by immunohistochemistry (IHC), immune cell markers (e.g. CD3, CD4, CD8; CSF1R, MHC-II) by cyclic immunofluorescence (cIF), and spatial transcriptomics (ST) using NanoString CosMx 1K panel.
- PPARγ and CC3 IHC images were analyzed using MIKAIA® software.
- Multiplexed cIF images were stitched and registered using ASHLAR[9]; cells were segmented using Cellpose-SAM[10], and marker positivity thresholds were determined using SCIMAP's NapariGater[11]. An in-house built 'Cyclic IF Analyses of Key Expressions' (CAKE) software was used for downstream analyses.
- ST data were processed using the NanoString AtoMx™ software.

FIGURE 1. NanoNail™ platform to generate *in-situ* pharmaco-phenotypic drug effects in humans

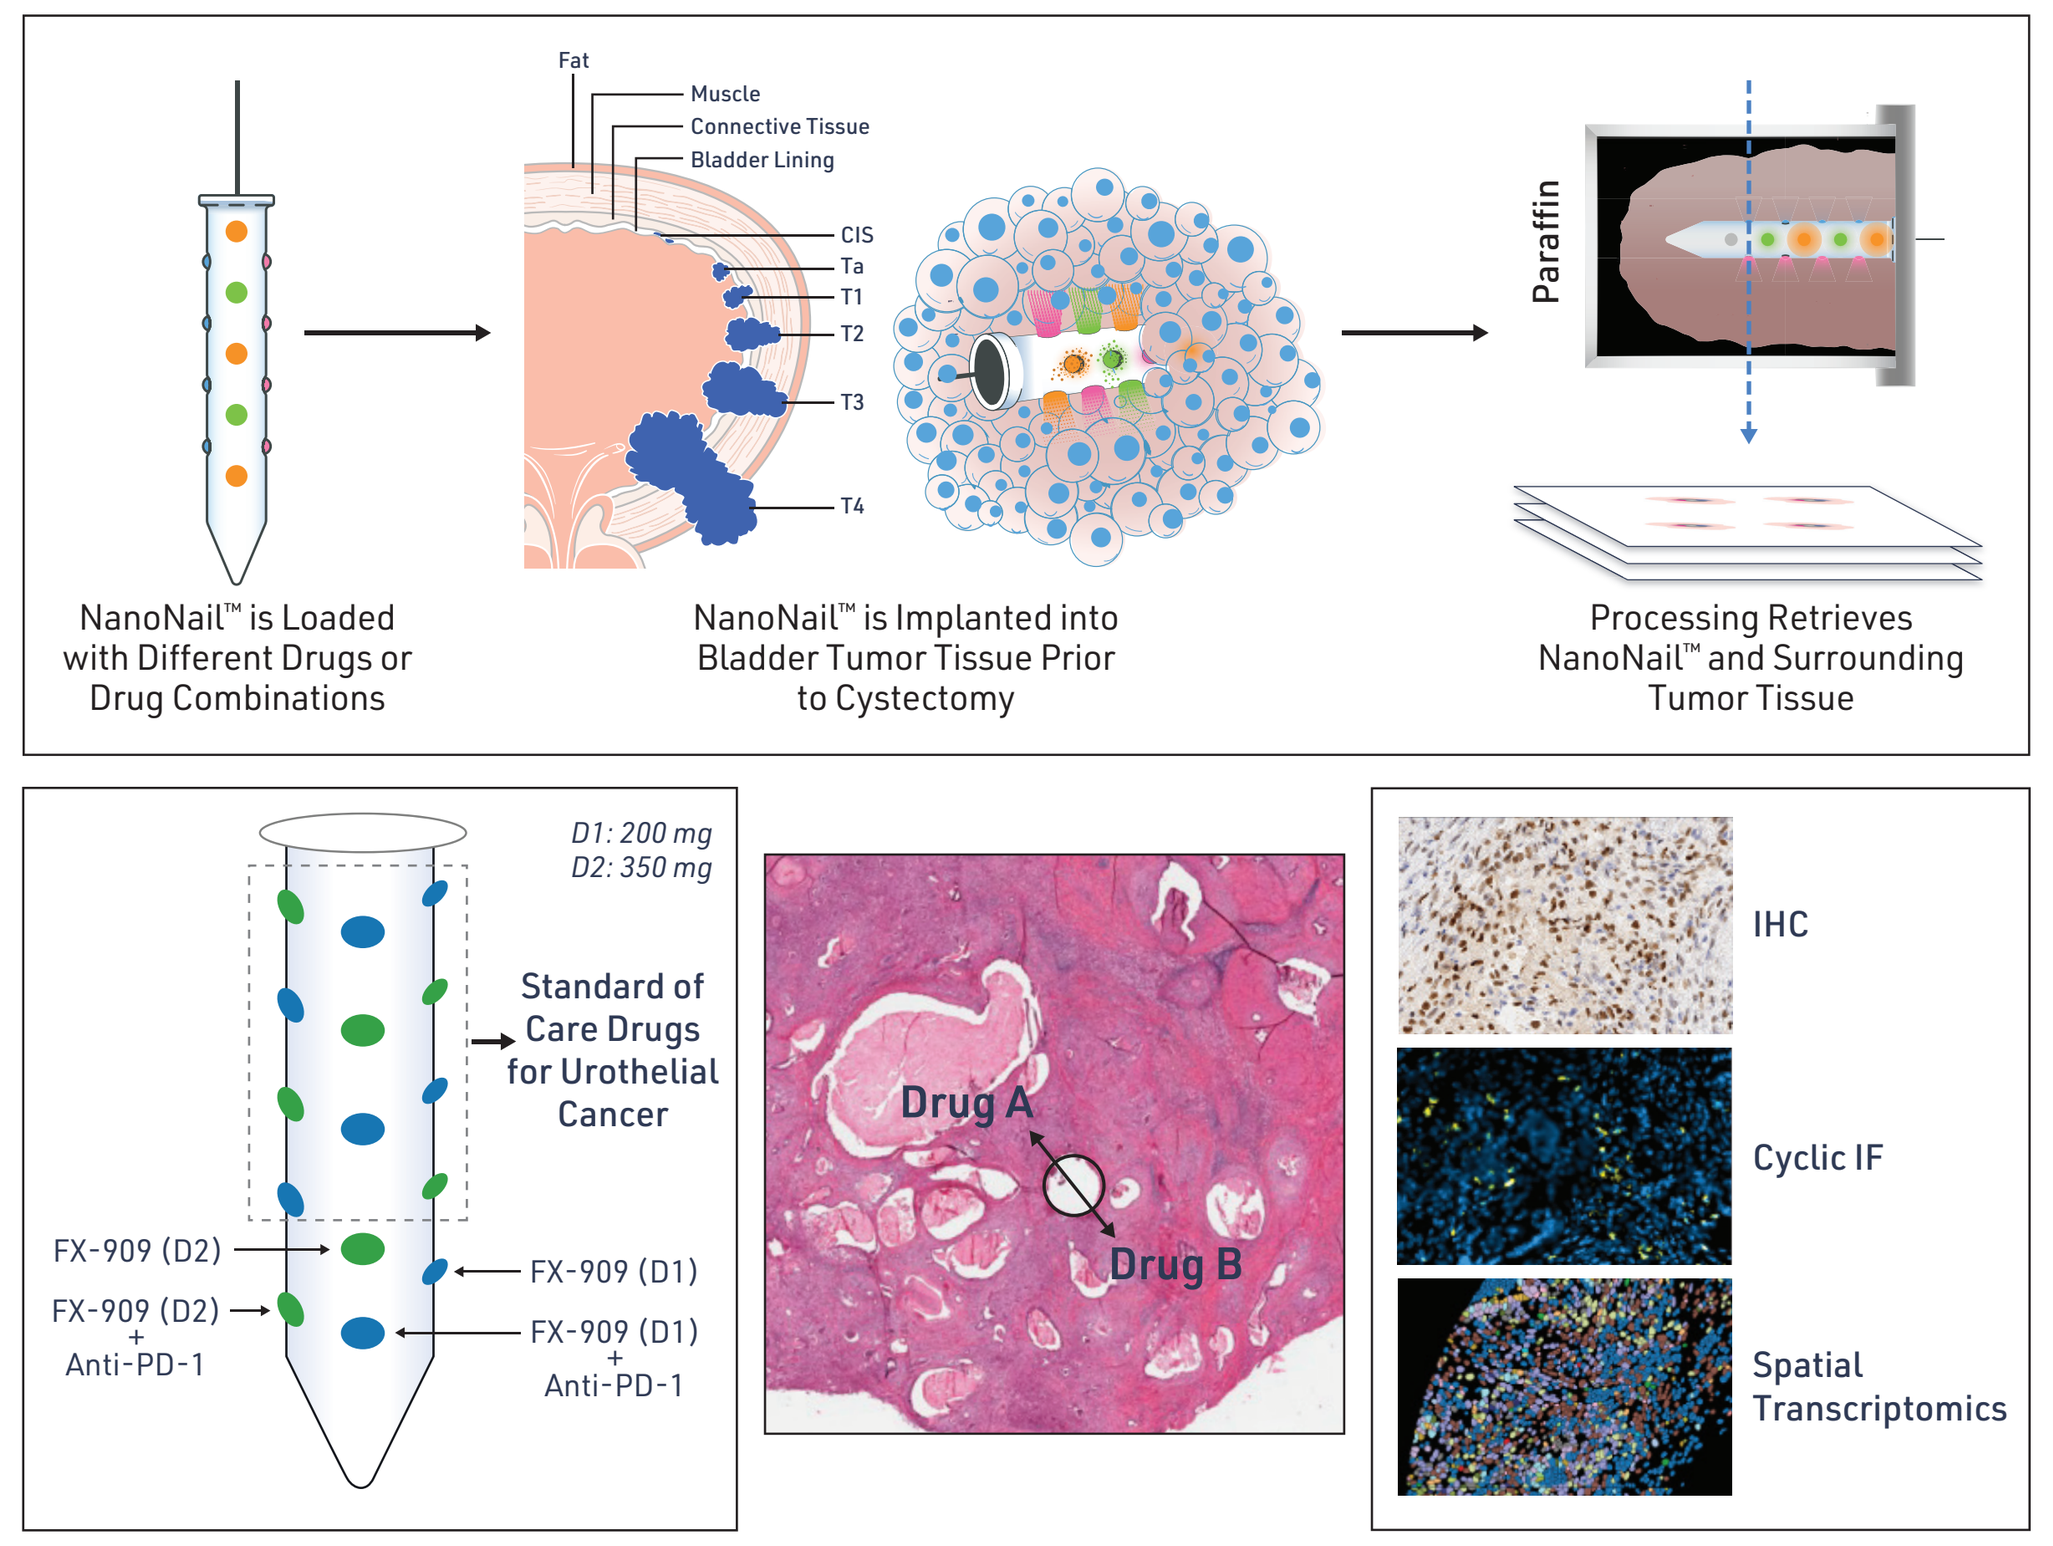

Fat
Muscle
Connective Tissue
Bladder Lining
CIS
Ta
T1
T2
T3
T4
Paraffin
NanoNail™ is Loaded with Different Drugs or Drug Combinations
NanoNail™ is Implanted into Bladder Tumor Tissue Prior to Cystectomy
Processing Retrieves NanoNail™ and Surrounding Tumor Tissue
D1: 200 mg
D2: 350 mg
Standard of Care Drugs for Urothelial Cancer
FX-909 (D2)
FX-909 (D2) + Anti-PD-1
FX-909 (D1)
FX-909 (D1) + Anti-PD-1
Drug A
Drug B
IHC
Cyclic IF
Spatial Transcriptomics

- Kibur Medical's NanoNail™ technology enables *in situ* tumor drug sensitivity profiling that supports early clinical testing of novel combinations in humans.

FIGURE 2. Patient characteristics: primary urothelial cancer

Patient ID	Diagnosis	Stage TNM	Prior Cancer Surgery	Prior Cancer Radiation Therapy	Prior Systemic Cancer Therapy	Sex	Race	Age	Duration of NanoNail™ Implantation
Pt1	Invasive adenocarcinoma with enteric differentiation	II T2, N0, M0	No	No	No	F	White	75	2 days
Pt2	Invasive high-grade carcinoma with extensive squamous differentiation	II T2, N0, M0	Yes	Yes	No	M	White	75	3 days

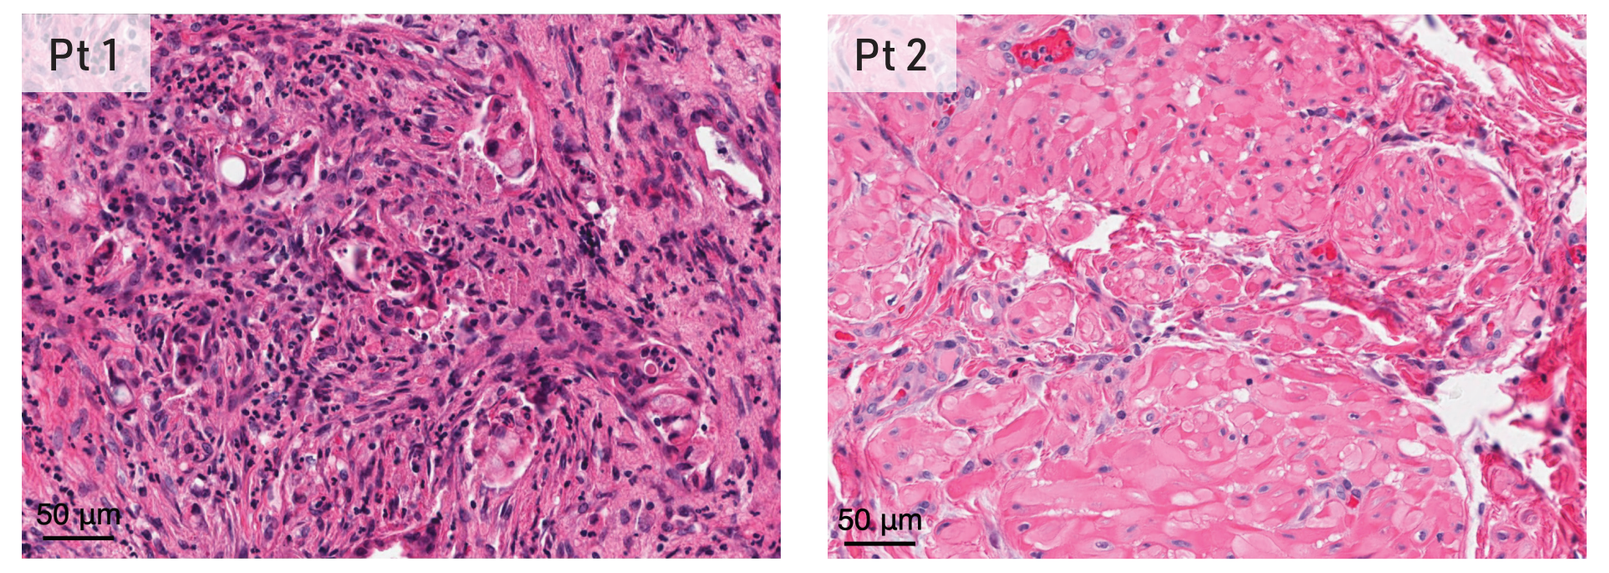

Pt 1
Pt 2
50 µm
50 µm

CONCLUSIONS

- In this Phase 0 study, FX-909 demonstrated intratumoral penetration and dose-dependent on-target PPARγ inhibition in primary bladder cancer through two complementary mechanisms:
 - Direct tumor cell killing via suppression of PPARγ-driven pathways and induction of apoptosis.
 - Reprogramming of immunosuppressive tumor-associated macrophages (TAMs) toward a pro-inflammatory phenotype with downstream dendritic cell (DC) activation and increased T cell infiltration.
- Modulation of other processes within the tumor microenvironment (TME) due to PPARγ inhibition is also observed, such as reduced angiogenesis gene signature in endothelial cells.
- Combined with anti-PD-1, FX-909 enhanced T cell infiltration and tumor cell killing, supporting further clinical evaluation of this combination in UC[4].

FX-909 unlocks additive anti-tumor activity with immune modulatory role via PPARγ inhibition

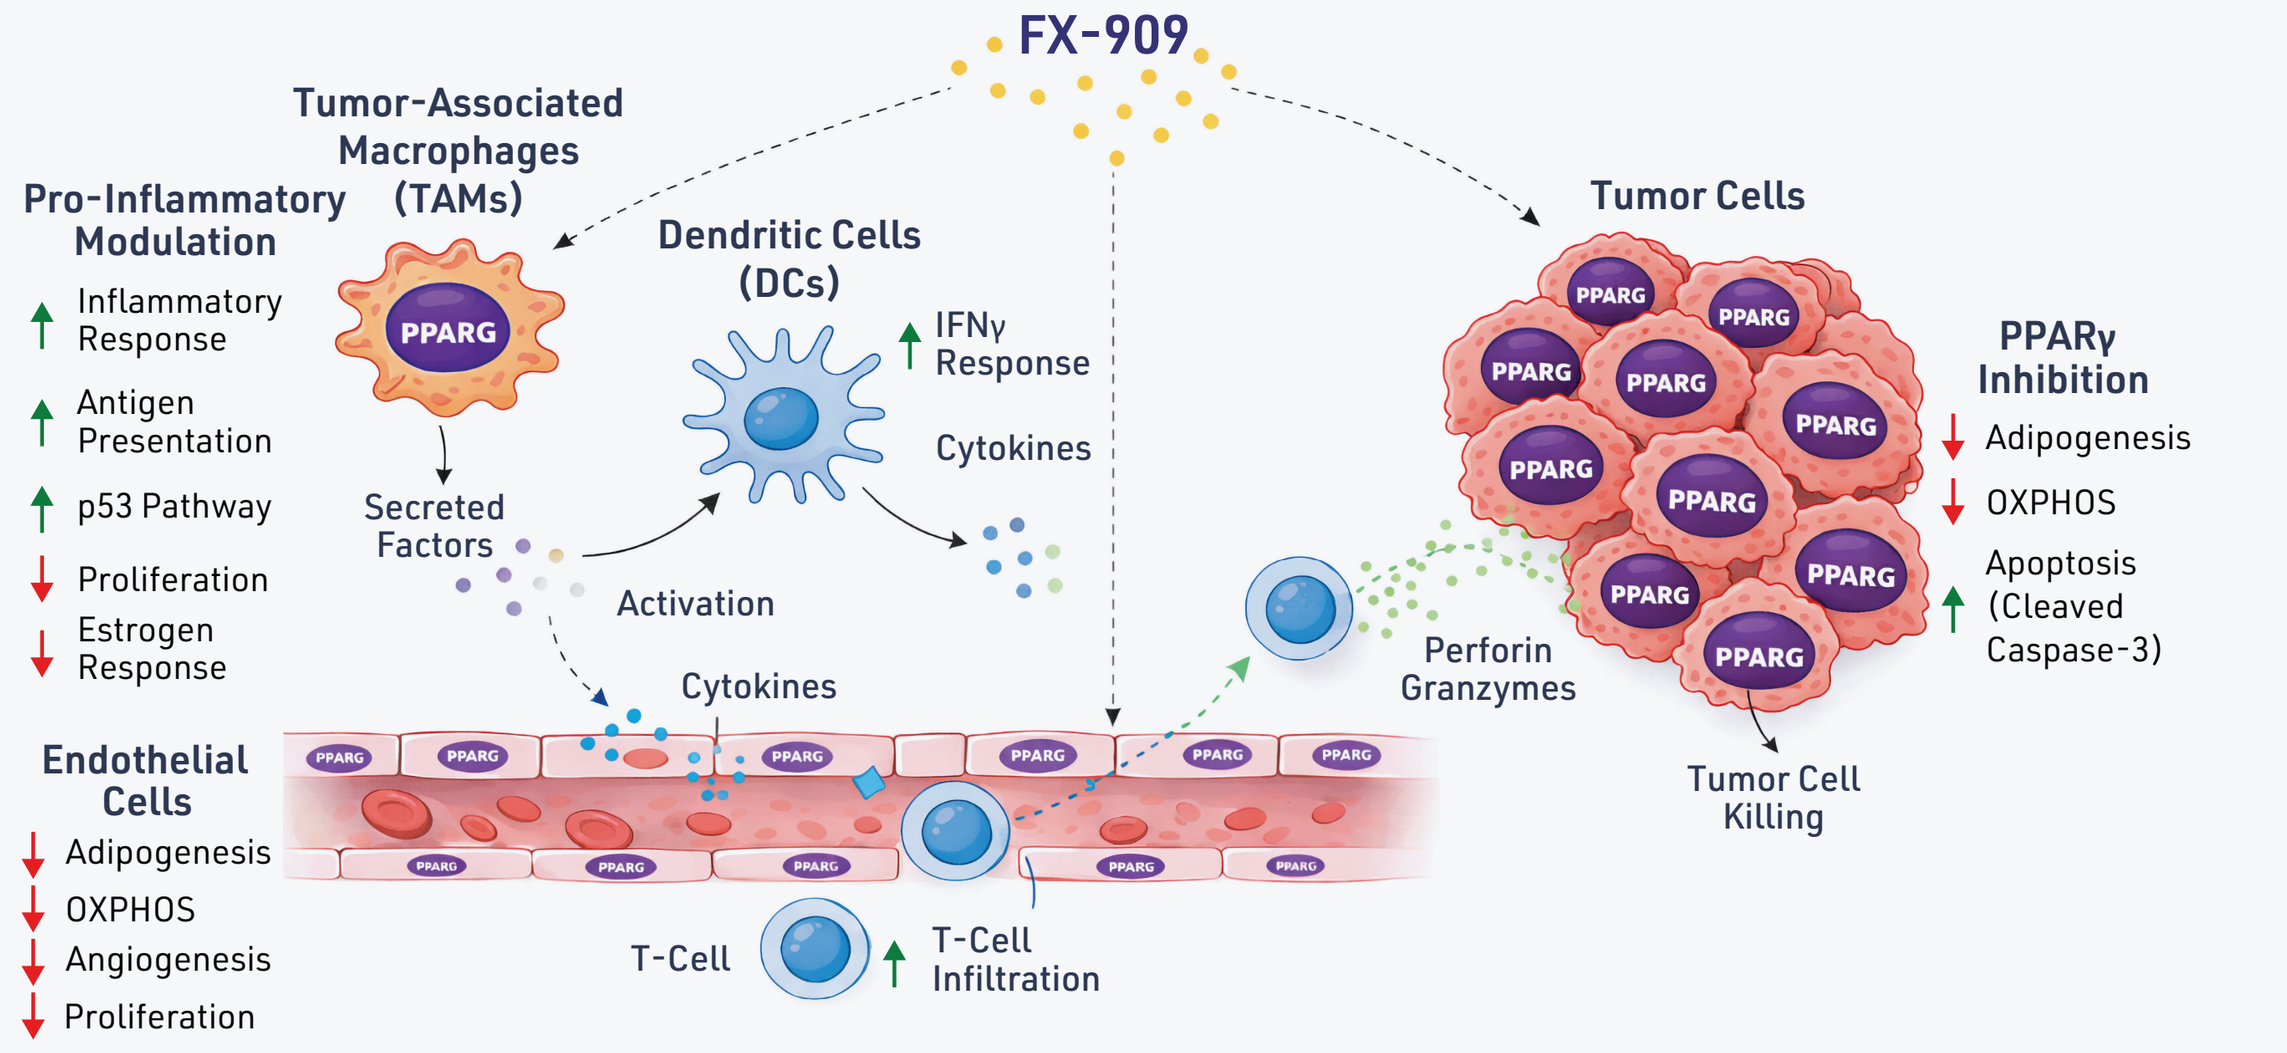

FX-909
Tumor-Associated Macrophages (TAMs)
Pro-Inflammatory Modulation
Inflammatory Response
Antigen Presentation
p53 Pathway
Proliferation
Estrogen Response
PPARG
Secreted Factors
Dendritic Cells (DCs)
IFNγ Response
Cytokines
Activation
Cytokines
Tumor Cells
PPARγ Inhibition
Adipogenesis
OXPHOS
Apoptosis (Cleaved Caspase-3)
Perforin Granzymes
Tumor Cell Killing
Endothelial Cells
Adipogenesis
OXPHOS
Angiogenesis
Proliferation
T-Cell
T-Cell Infiltration

RESULTS

FIGURE 3. Assessment of PPARγ and Cleaved Caspase-3 (CC3) expression

3A. Patient 1: ~20% tumor content

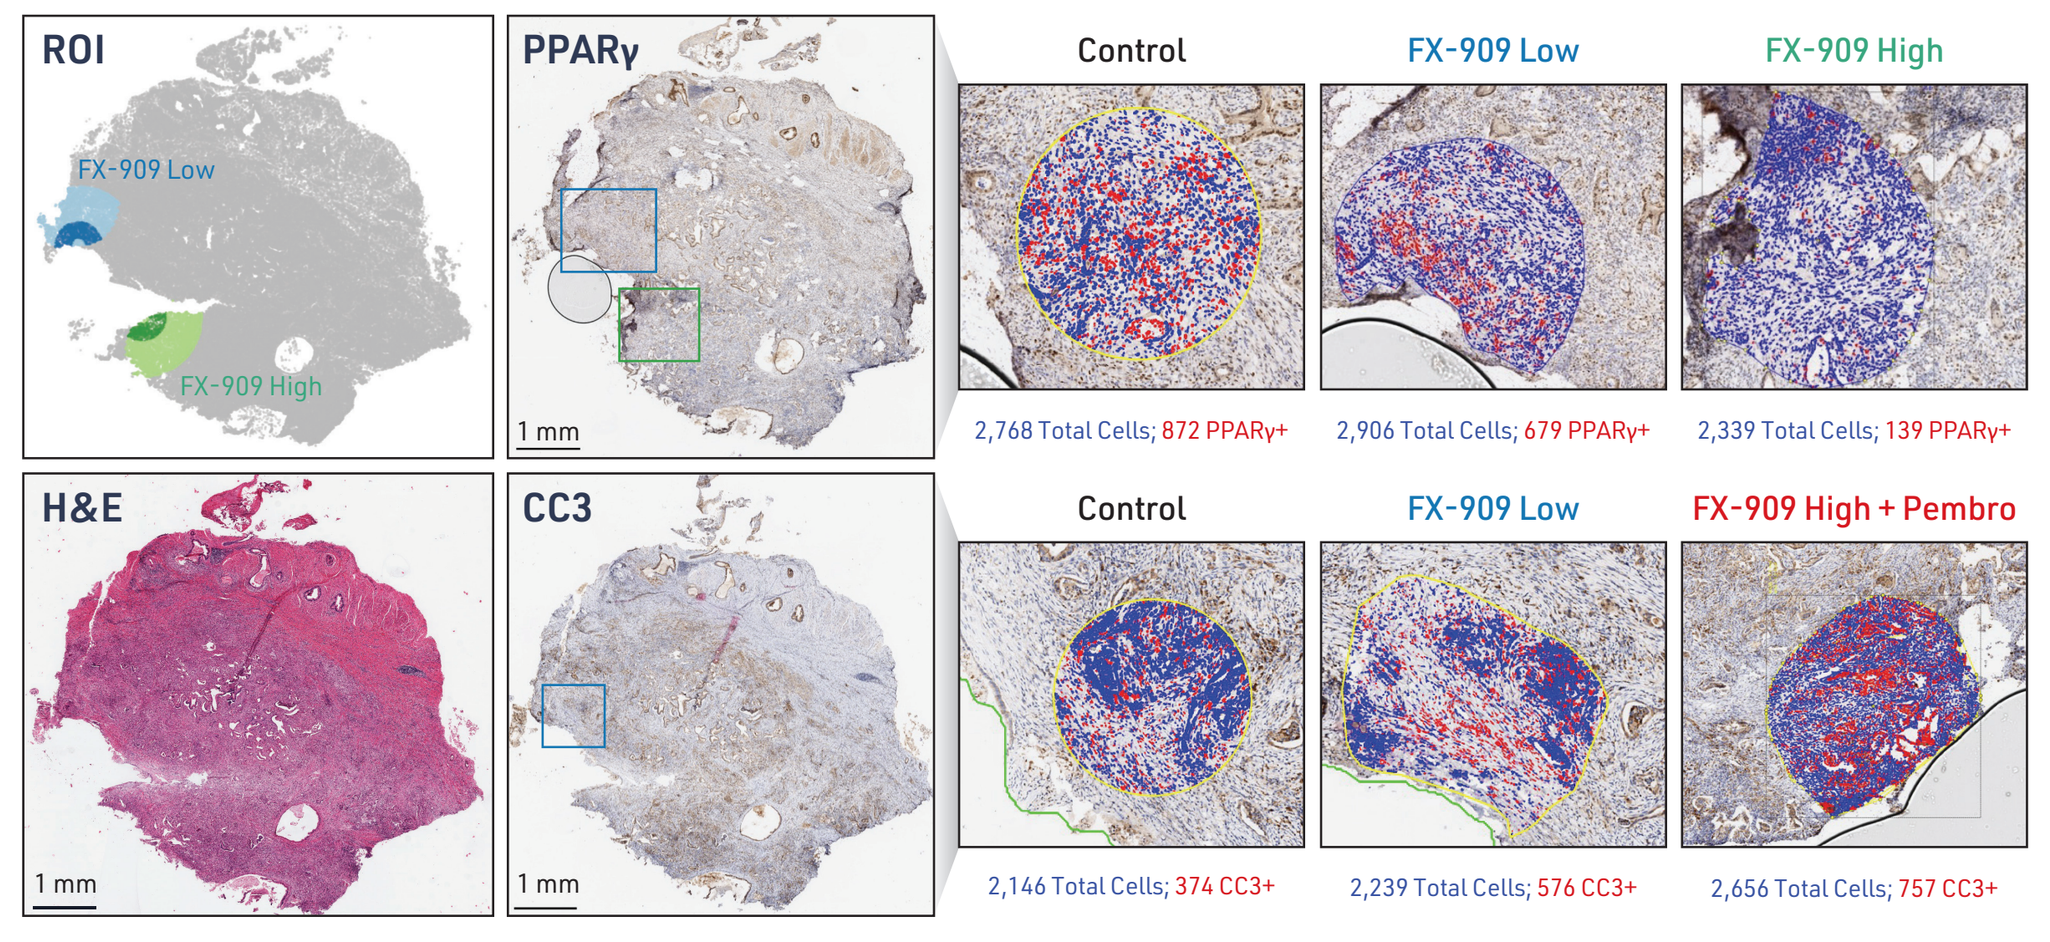

ROI
FX-909 Low
FX-909 High
PPARγ
Control
FX-909 Low
FX-909 High
2,768 Total Cells; 872 PPARγ+
2,906 Total Cells; 679 PPARγ+
2,339 Total Cells; 139 PPARγ+
H&E
CC3
Control
FX-909 Low
FX-909 High + Pembro
2,146 Total Cells; 374 CC3+
2,239 Total Cells; 576 CC3+
2,656 Total Cells; 757 CC3+
1 mm

Dark Bands: Regions with highest FX-909 exposure (100-400 µm from NanoNail)

3B. Patient 2: ~9% tumor content

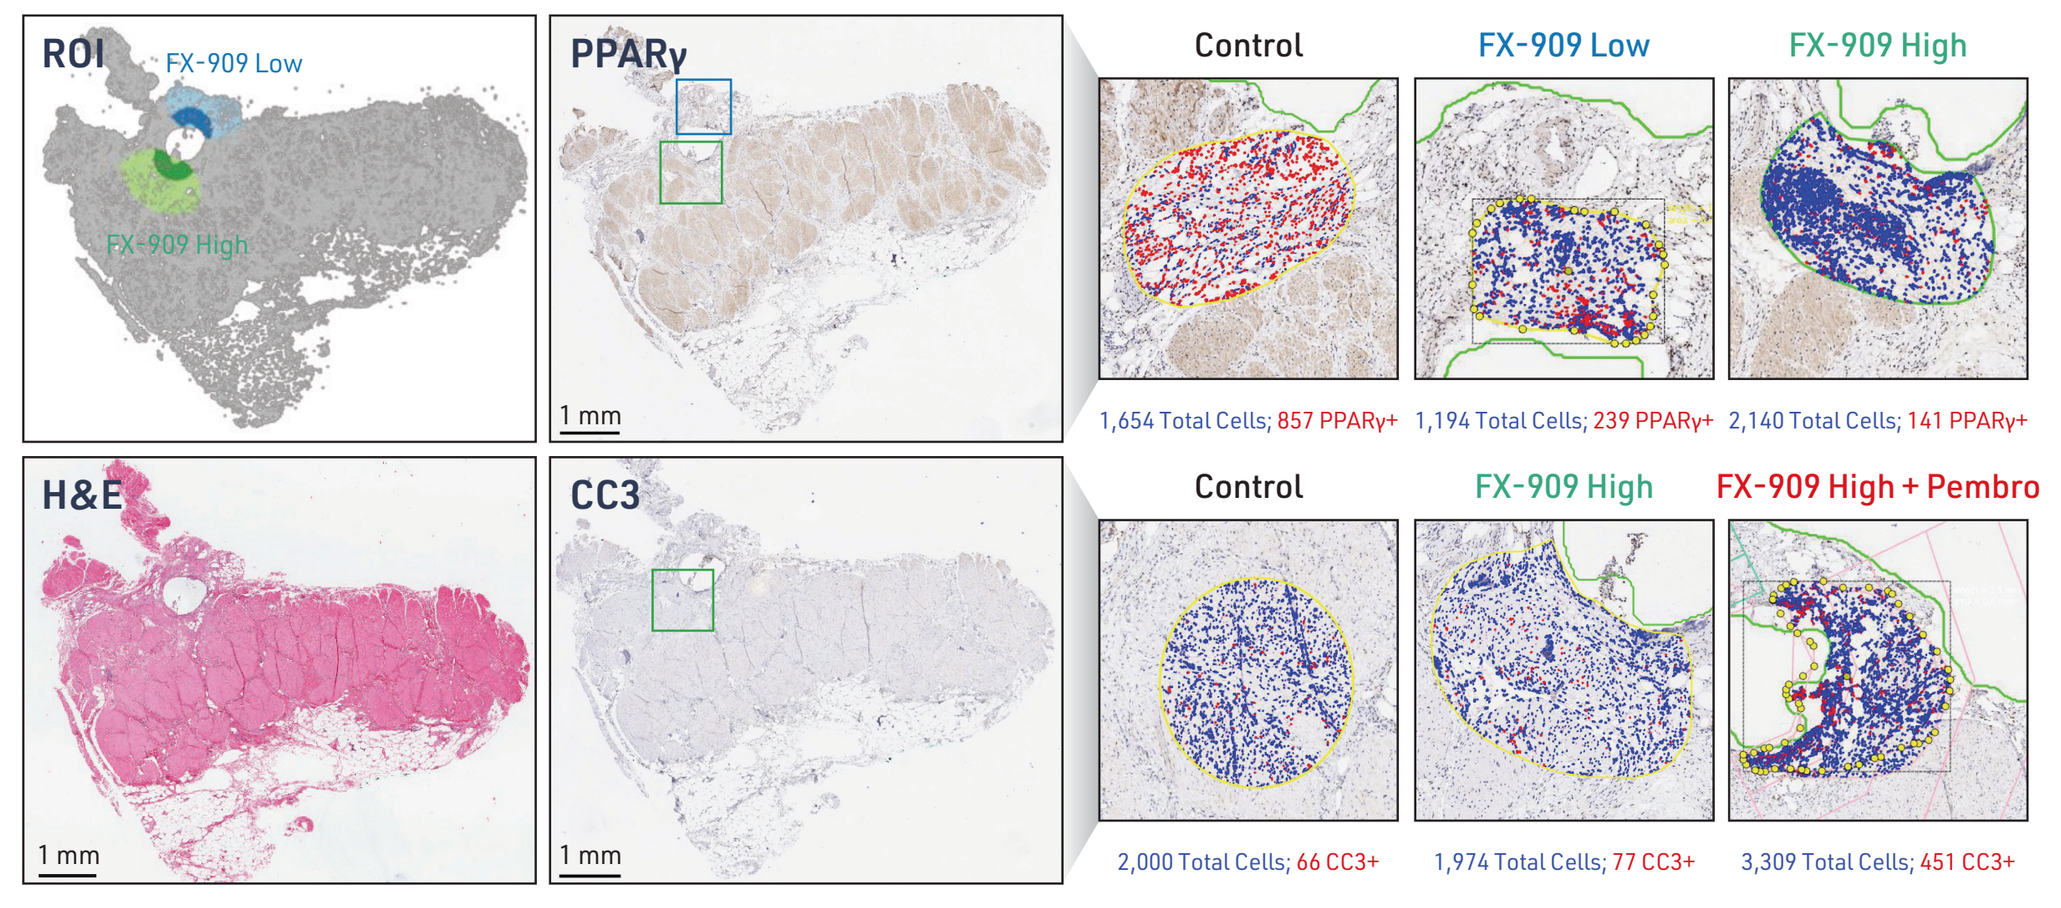

ROI
FX-909 Low
FX-909 High
PPARγ
Control
FX-909 Low
FX-909 High
1,654 Total Cells; 857 PPARγ+
1,194 Total Cells; 239 PPARγ+
2,140 Total Cells; 141 PPARγ+
H&E
CC3
Control
FX-909 High
FX-909 High + Pembro
2,000 Total Cells; 66 CC3+
1,974 Total Cells; 77 CC3+
3,309 Total Cells; 451 CC3+
1 mm

Dark Bands: Regions with highest FX-909 exposure (100-400 µm from NanoNail)

3C. *PPARG* mRNA expression by cell type and patient

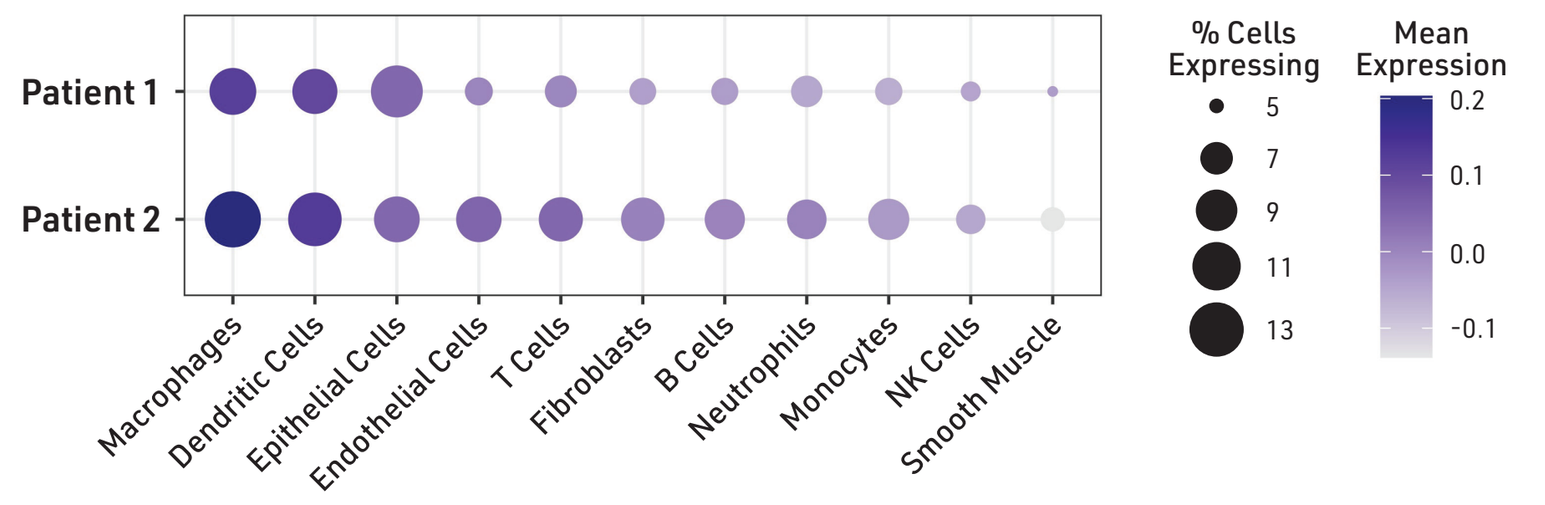

Patient 1
Patient 2
Macrophages
Dendritic Cells
Epithelial Cells
Endothelial Cells
T Cells
Fibroblasts
B Cells
Neutrophils
Monocytes
NK Cells
Smooth Muscle
% Cells Expressing
5
7
9
11
13
Mean Expression
0.2
0.1
0.0
-0.1

- *PPARG* is predominantly expressed in macrophages, dendritic cells (DCs), epithelial (tumor) cells and endothelial cells.
- Caveat: *PPARG* mRNA is likely under-detected in CosMx 1K assay.

3D. FX-909 reduces PPARγ protein expressing cells in a dose-dependent manner

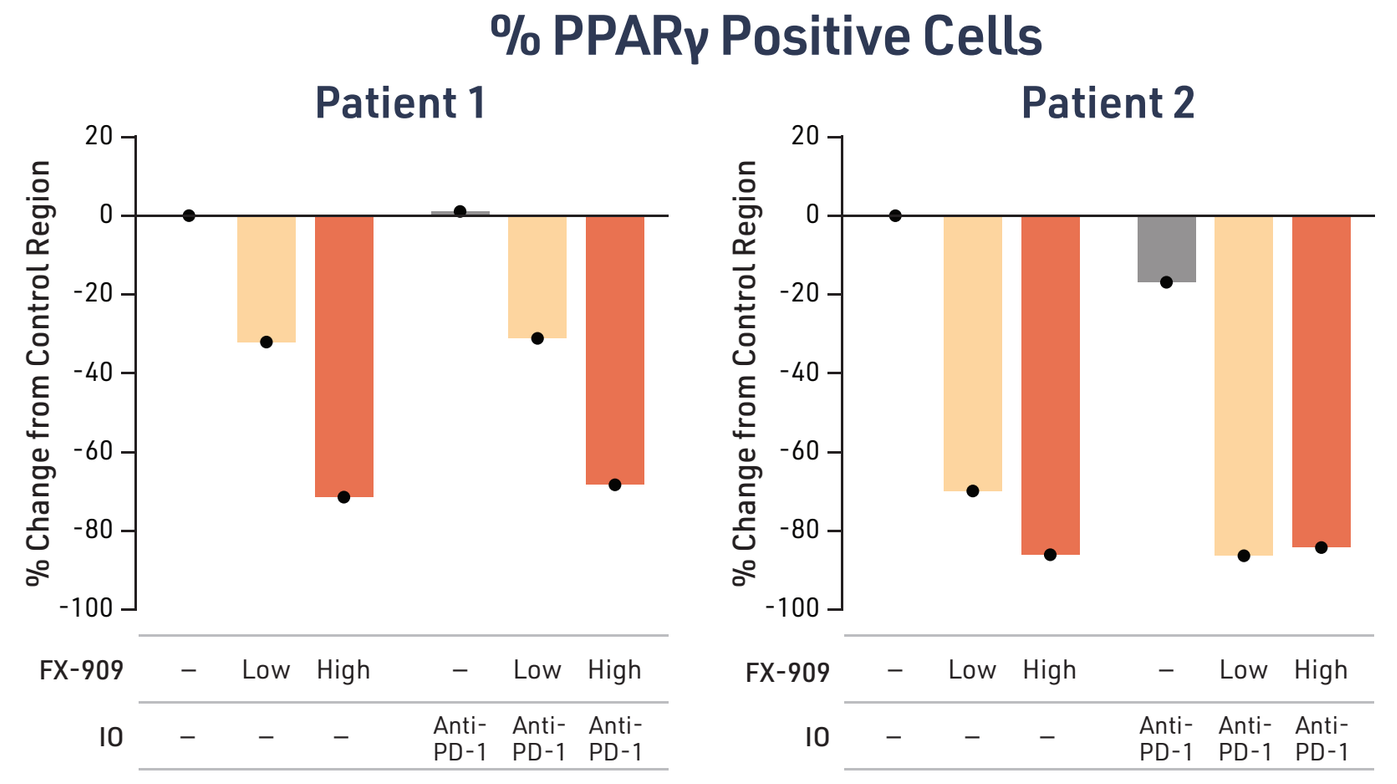

% PPARγ Positive Cells
Patient 1
Patient 2
% Change from Control Region
FX-909
IO
Low
High
Anti-PD-1

- In Pt 1, most of PPARγ+ cells are epithelial tumor cells, hence the more pronounced dose-response reflecting FX-909 induced tumor cell killing
- In Pt 2, most of PPARγ+ cells are macrophages and DCs with unknown response to FX-909 treatment

3E. FX-909 increases apoptotic index (% CC3+ cells) alone and in combination with Anti-PD-1

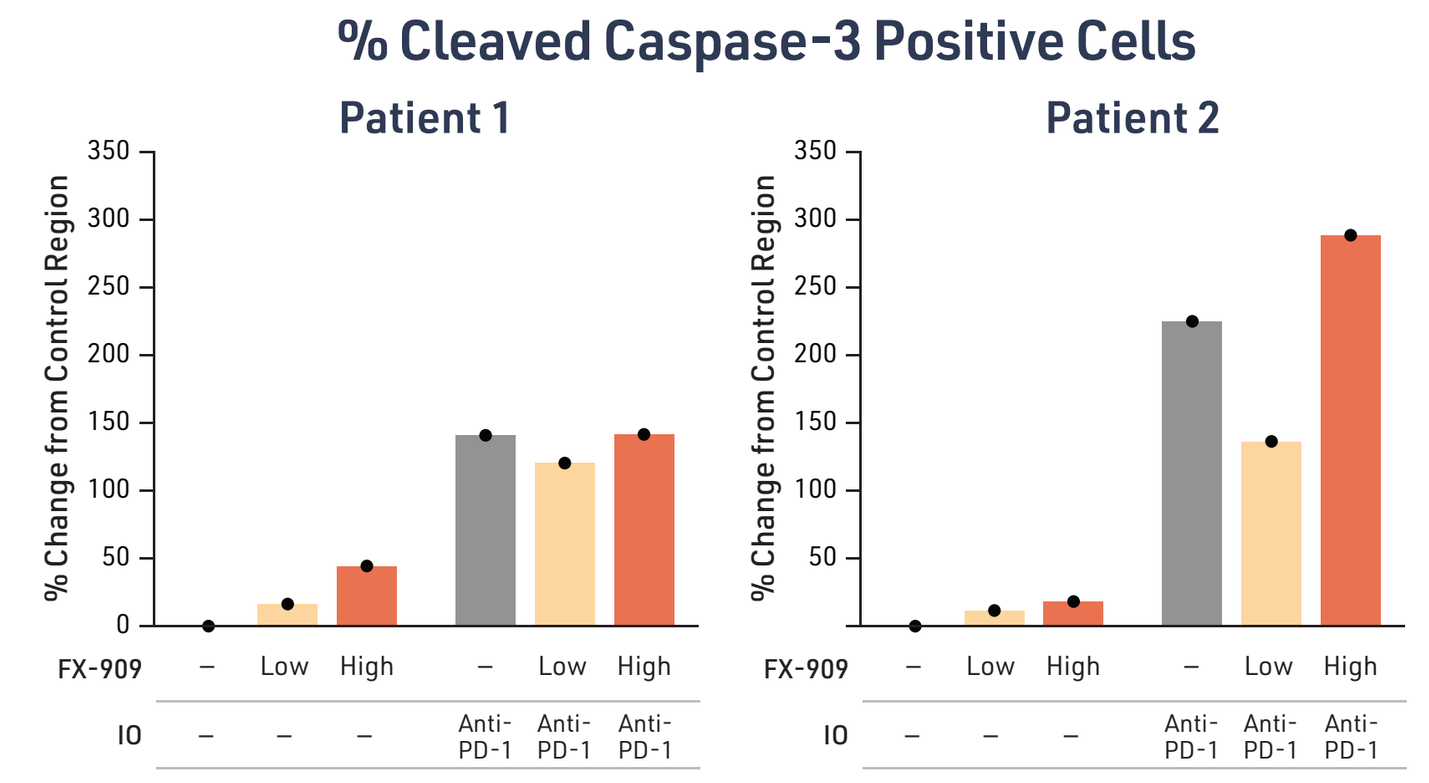

% Cleaved Caspase-3 Positive Cells
Patient 1
Patient 2
% Change from Control Region
FX-909
IO
Low
High
Anti-PD-1

FIGURE 4. Determination of distance and concentration-dependent effects of FX-909

4A. Assessment of FX-909 tissue penetration (MALDI-IMS; Pt1)

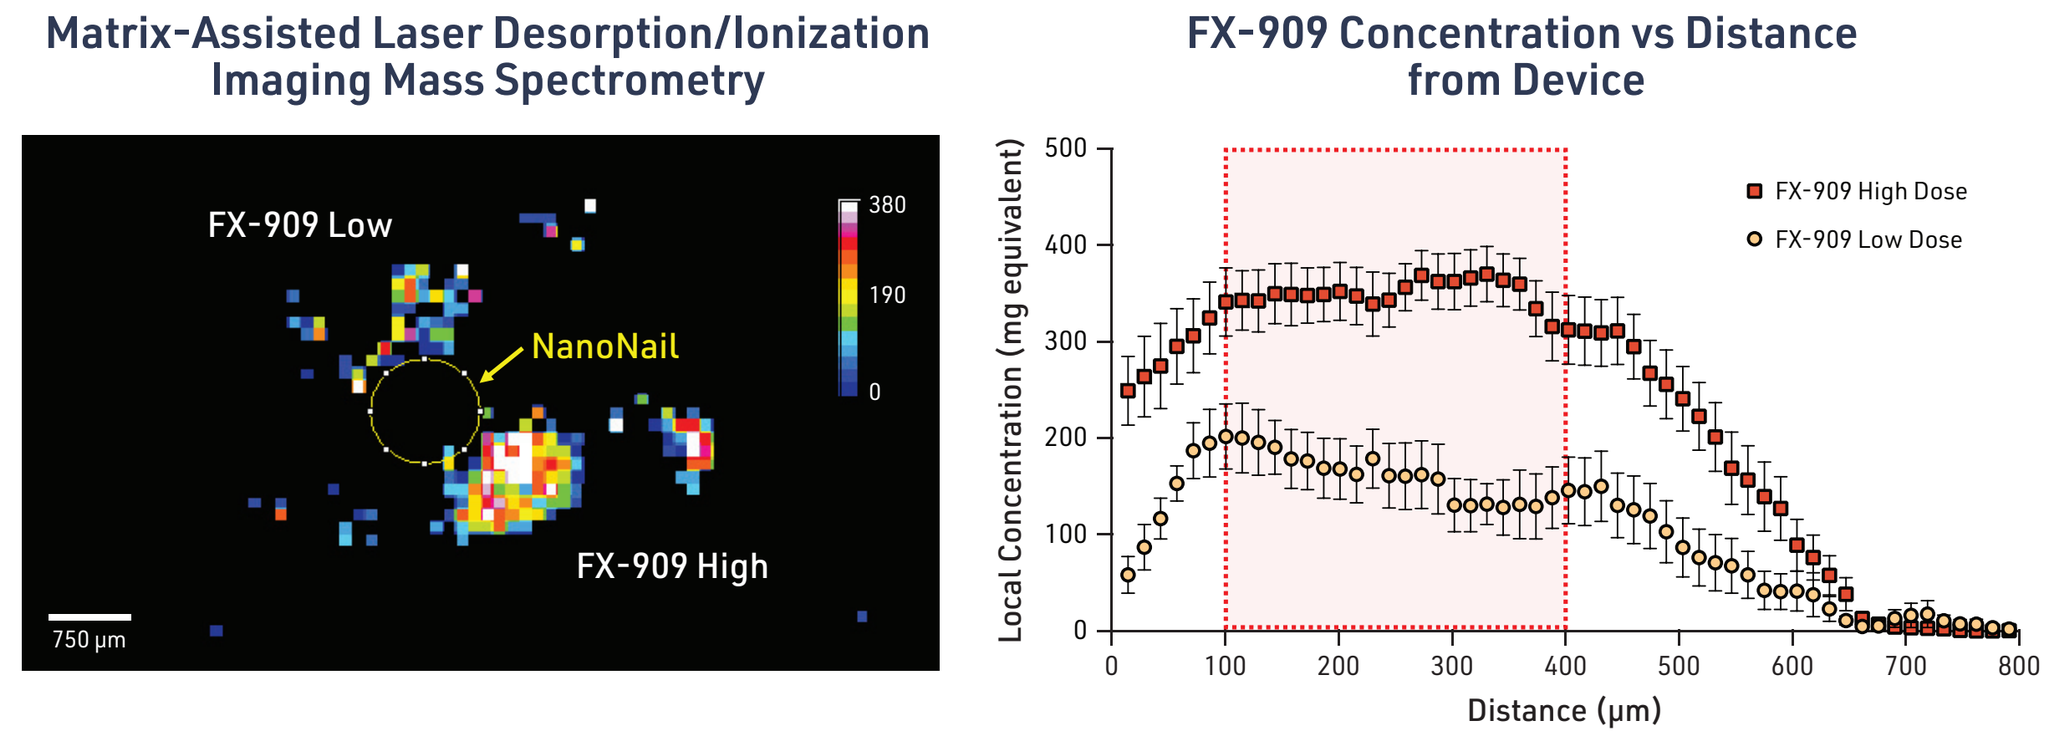

Matrix-Assisted Laser Desorption/Ionization Imaging Mass Spectrometry
FX-909 Low
NanoNail
FX-909 High
750 µm
FX-909 Concentration vs Distance from Device
Local Concentration (mg equivalent)
Distance (µm)
FX-909 High Dose
FX-909 Low Dose

- FX-909 tissue penetration showed a dose- and distance- dependent concentration gradient, highest near the device-tissue interface.
- FX-909 concentrations plateaued at 100-400 µm from the device, with similar histomorphology in exposed and unexposed control-regions.

4B. Pathway scoring using spatial transcriptomics data

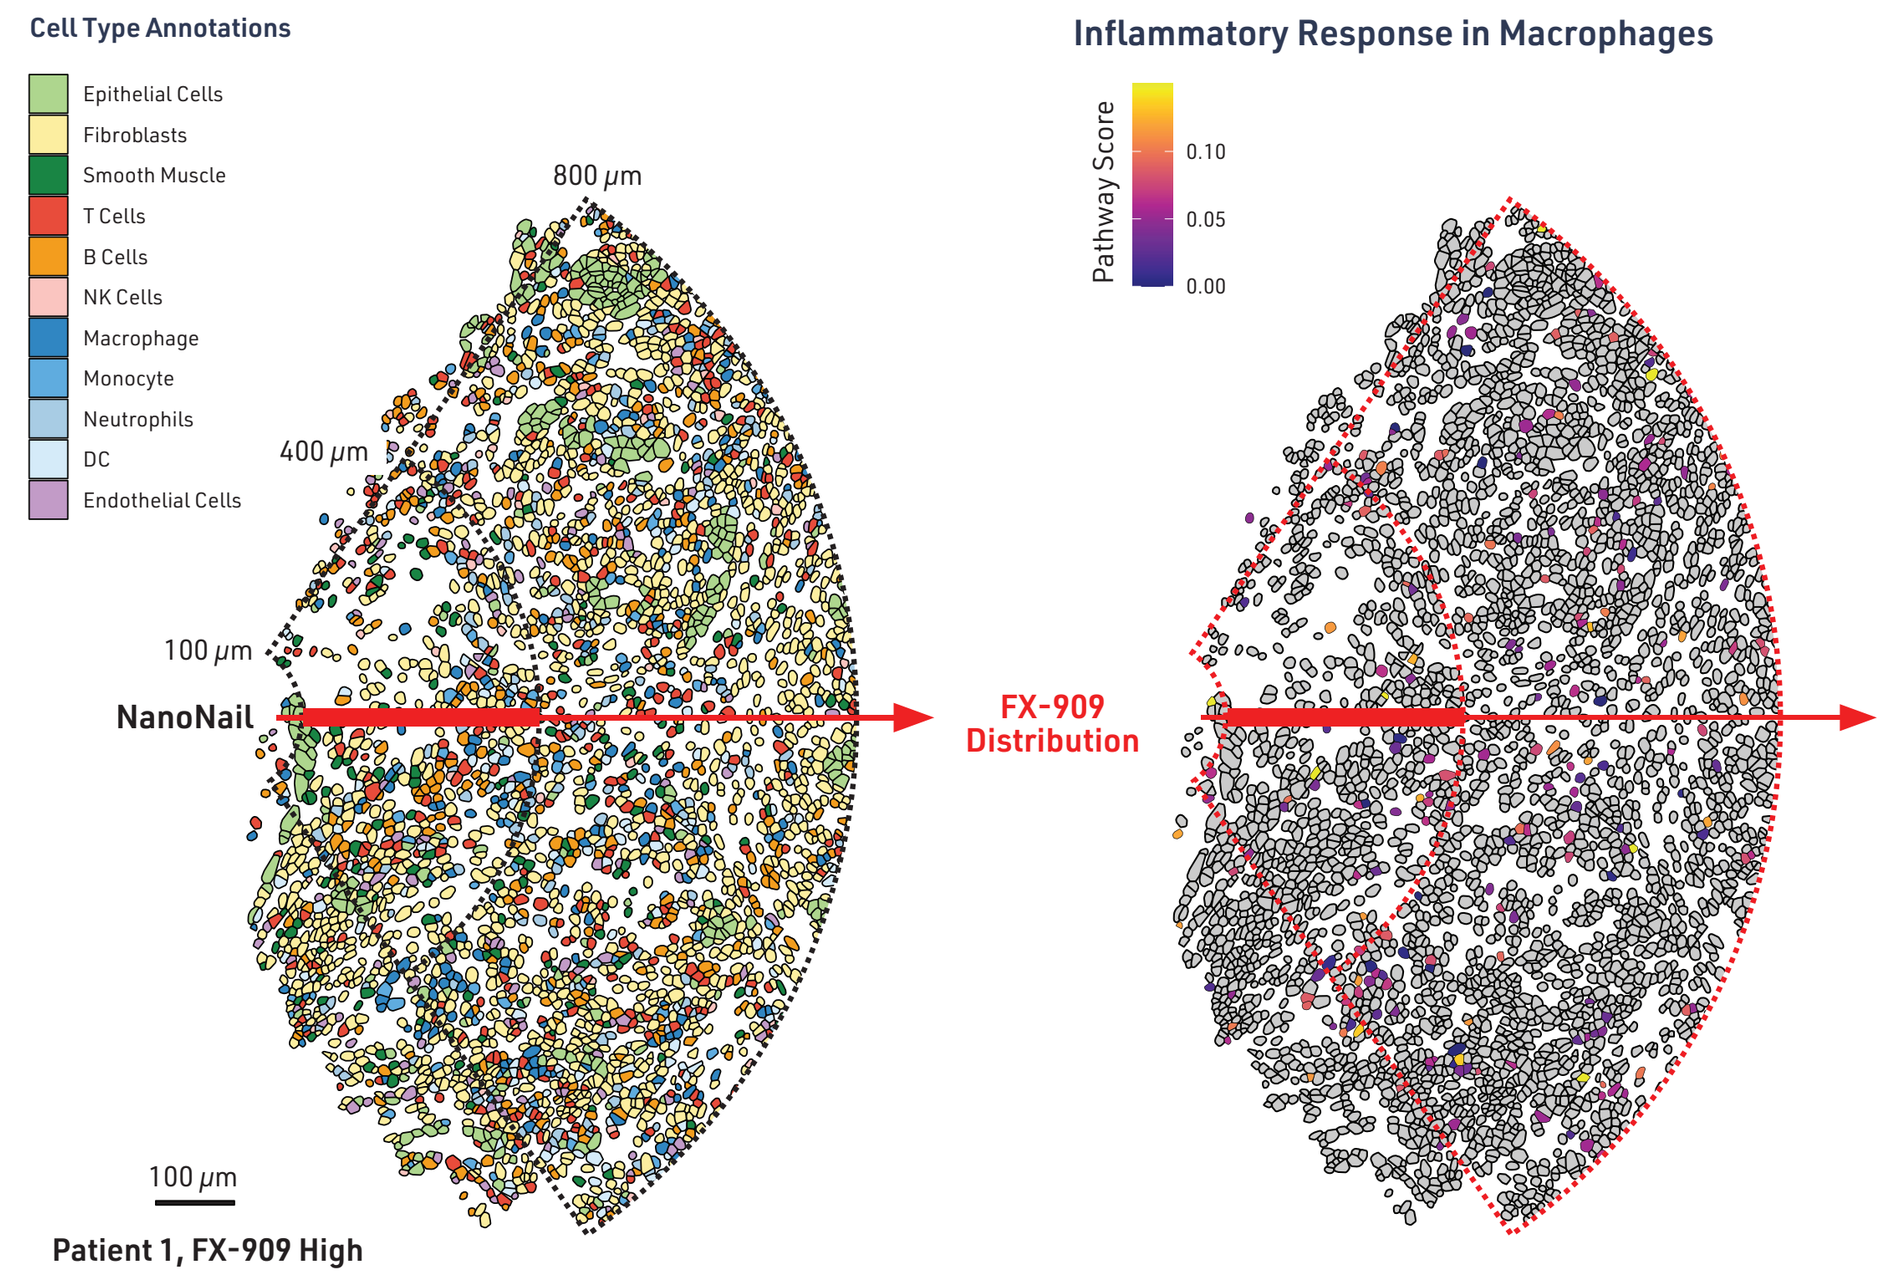

Cell Type Annotations
Epithelial Cells
Fibroblasts
Smooth Muscle
T Cells
B Cells
NK Cells
Macrophage
Monocyte
Neutrophils
DC
Endothelial Cells
800 µm
400 µm
100 µm
NanoNail
FX-909 Distribution
Patient 1, FX-909 High
Inflammatory Response in Macrophages
Pathway Score

- Cells were segmented using DAPI, PanCK, CK8/18, pan-membrane, and CD45 IF staining.
- Following cell-level QC and filtering, cells were annotated using a combination of unsupervised and supervised methods:
 - **The majority of tumor cells were annotated as "Epithelial Cells"**
- Per-cell pathway scores were calculated for Hallmark pathways with more than three shared genes in the CosMx 1K panel using *decoupleR's* implementation of the *AUCell* algorithm.
- For each cell type, pathway scores were averaged across cells within each distance bin.

FIGURE 5. Distance-dependent effects of FX-909

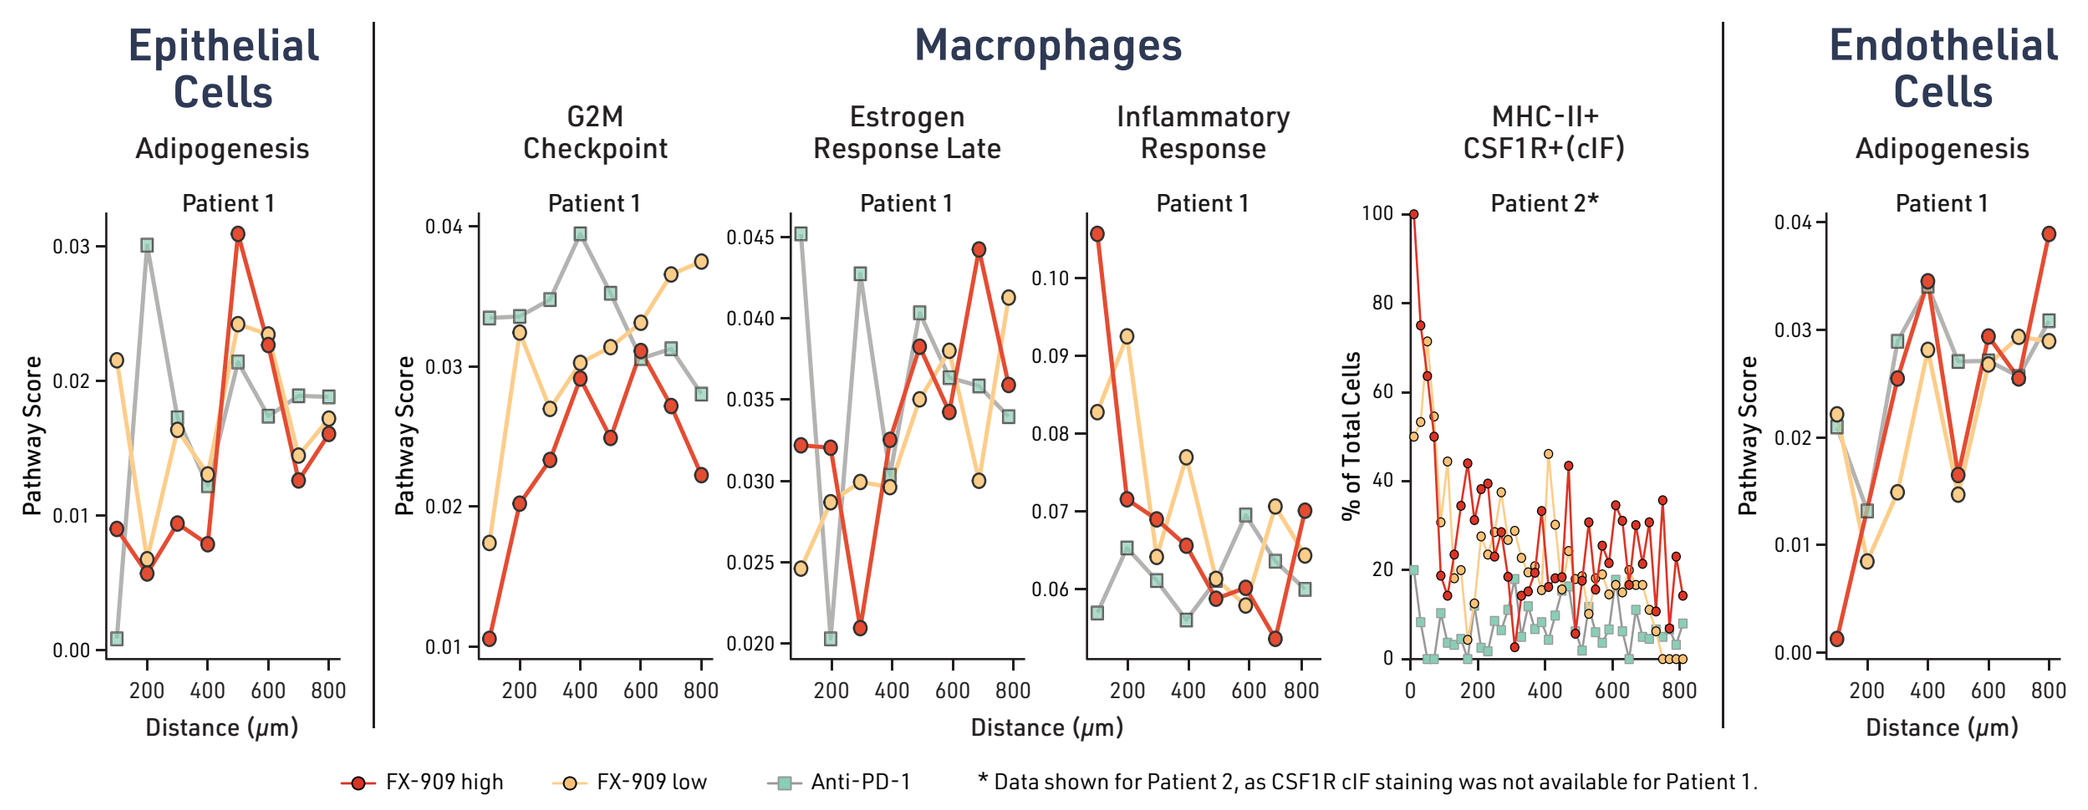

Epithelial Cells
Adipogenesis
Macrophages
G2M Checkpoint
Estrogen Response Late
Inflammatory Response
MHC-II+ CSF1R+(cIF)
Endothelial Cells
Adipogenesis
Patient 1
Patient 2*
Pathway Score
% of Total Cells
Distance (µm)
FX-909 high
FX-909 low
Anti-PD-1
* Data shown for Patient 2, as CSF1R cIF staining was not available for Patient 1.

FIGURE 6. Integration of MALDI and spatial transcriptomics data reveals pathways modulated by FX-909 in a concentration-dependent manner

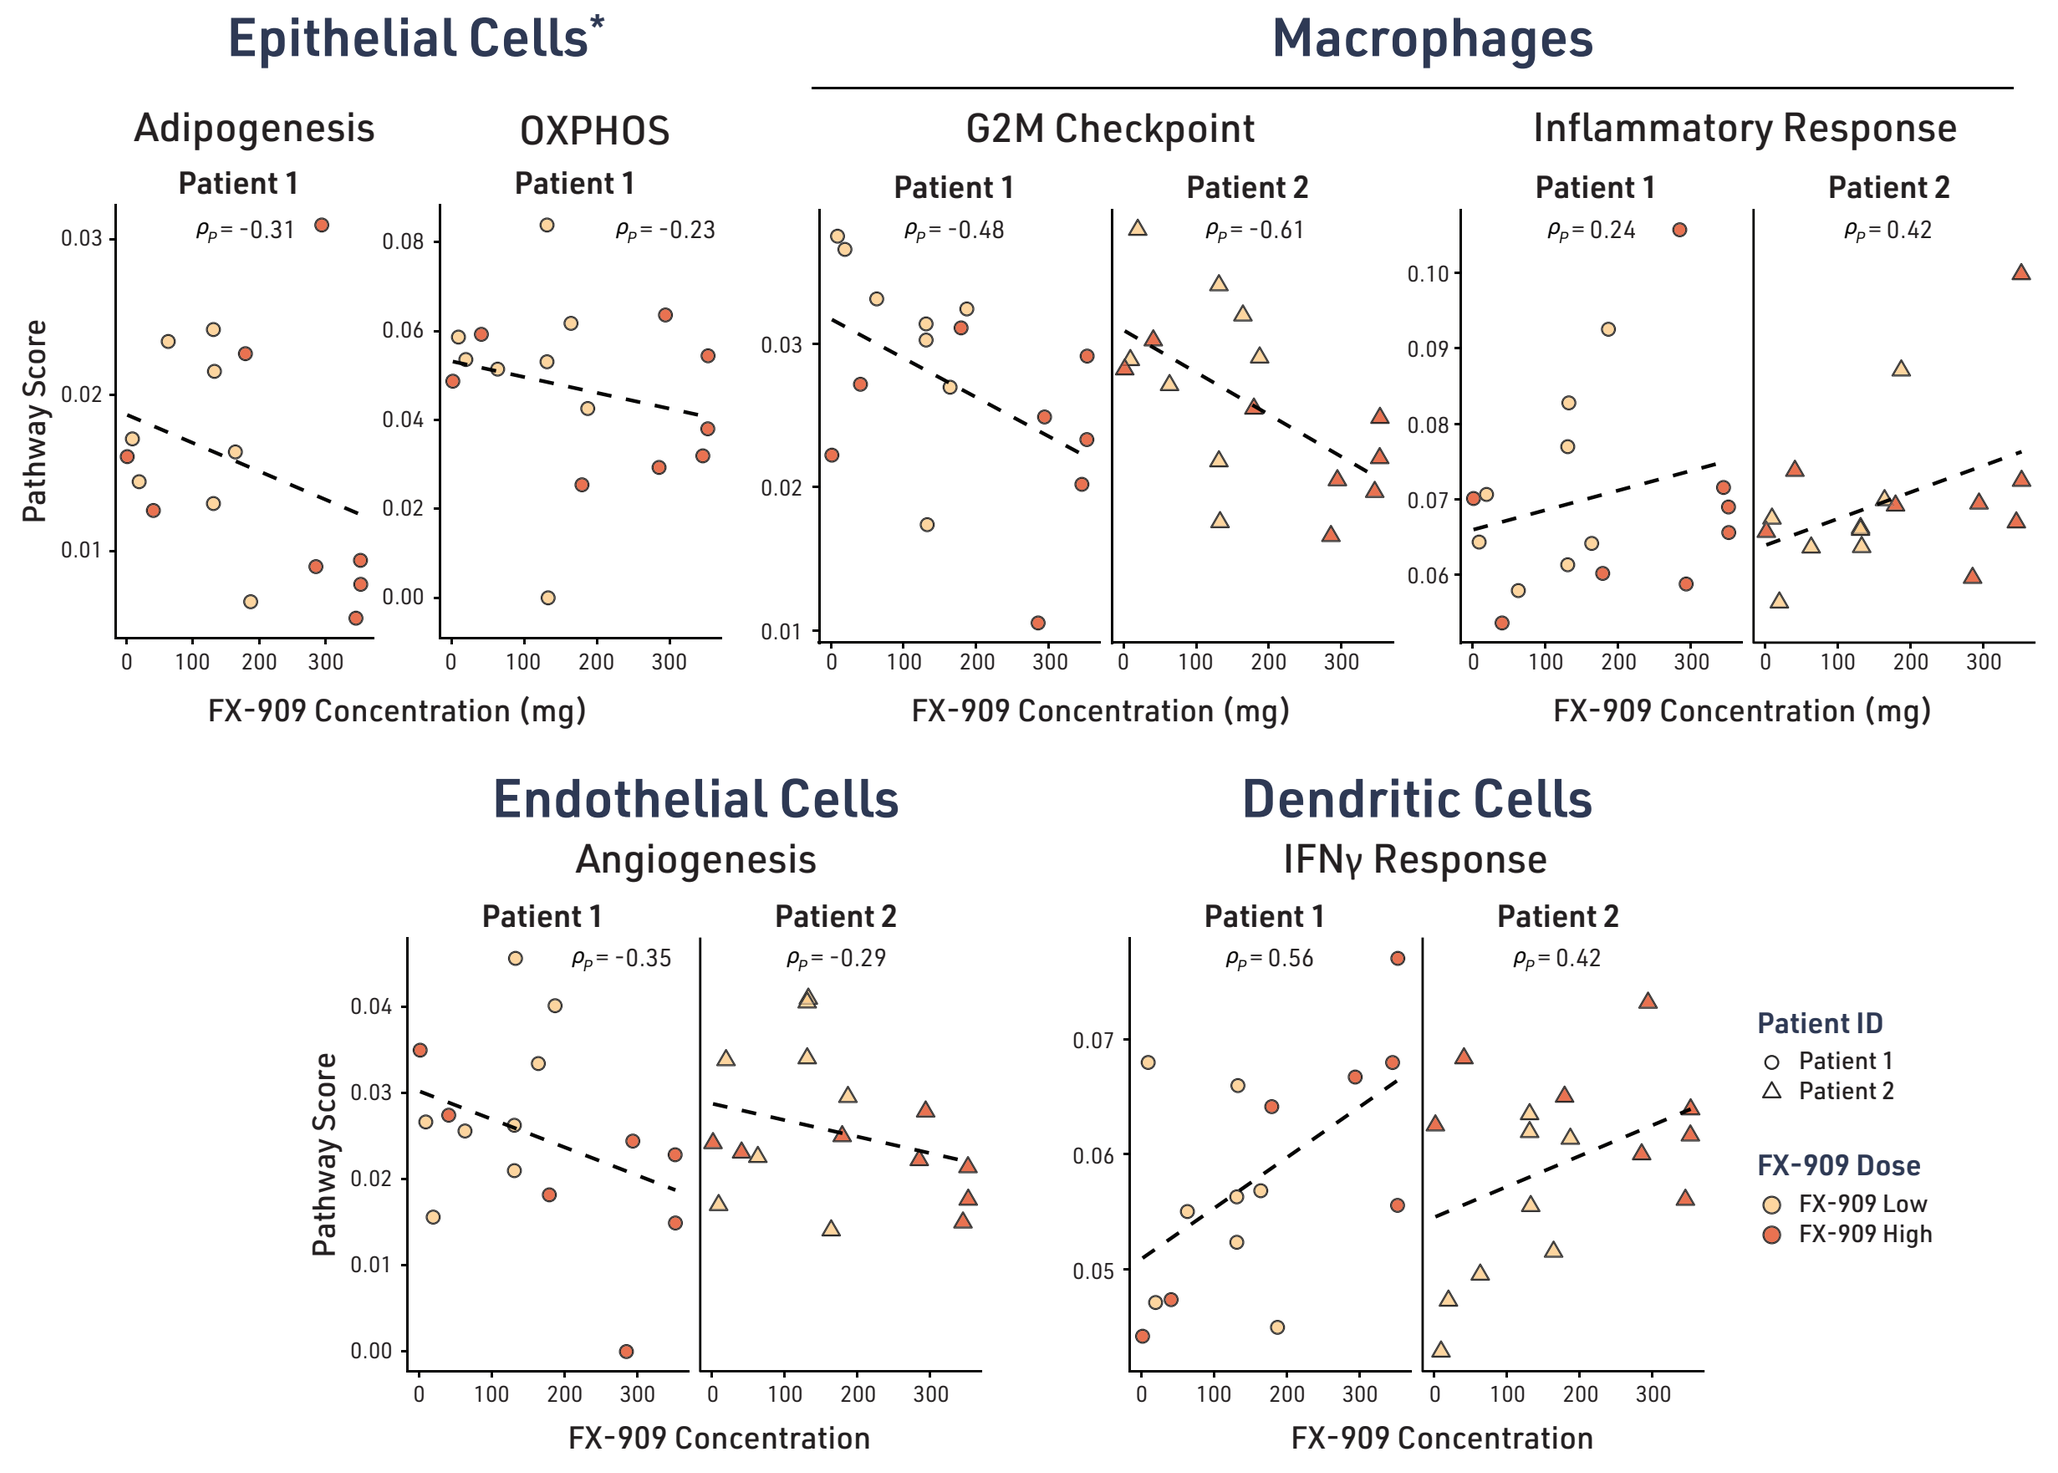

Epithelial Cells*
Adipogenesis
OXPHOS
Macrophages
G2M Checkpoint
Inflammatory Response
Endothelial Cells
Angiogenesis
Dendritic Cells
IFNγ Response
Patient 1
Patient 2
Pathway Score
FX-909 Concentration (mg)
FX-909 Concentration
Patient ID
FX-909 Dose
FX-909 Low
FX-909 High

- Linear regression was performed on FX-909 concentrations, determined from MALDI-IMS at both high and low dose levels, versus averaged cell-level pathway expression scores binned by distance from NanoNail™.

Cell Type	Hallmark Pathway	Patient	Pearson ρ
Epithelial Tumor Cells*	Adipogenesis	Pt1	-0.31
	Oxidative Phosphorylation	Pt1	-0.23
Macrophages	G2M Checkpoint	Pt1	-0.48
		Pt2	-0.61
	Mitotic Spindle	Pt1	-0.38
		Pt2	-0.47
	Estrogen Response Early	Pt1	-0.34
		Pt2	-0.47
	Estrogen Response Late	Pt1	-0.44
		Pt2	-0.29
	Inflammatory Response	Pt1	0.24
		Pt2	0.42
	p53 Pathway	Pt1	0.37
		Pt2	0.33
Endothelial Cells	Oxidative Phosphorylation	Pt1	-0.32
		Pt2	-0.38
	Adipogenesis	Pt1	-0.36
		Pt2	-0.23
	E2F Targets	Pt1	-0.25
		Pt2	-0.45
	Angiogenesis	Pt1	-0.35
		Pt2	-0.29
Dendritic Cells	IFNγ Response	Pt1	0.56
		Pt2	0.42

* Number of epithelial tumor cells in Pt2 spatial transcriptomics data was too low to be analyzed.

FIGURE 7. FX-909 increases CD8+ T cell infiltration in the TME alone and in combination with Anti-PD-1 (Cyclic IF)

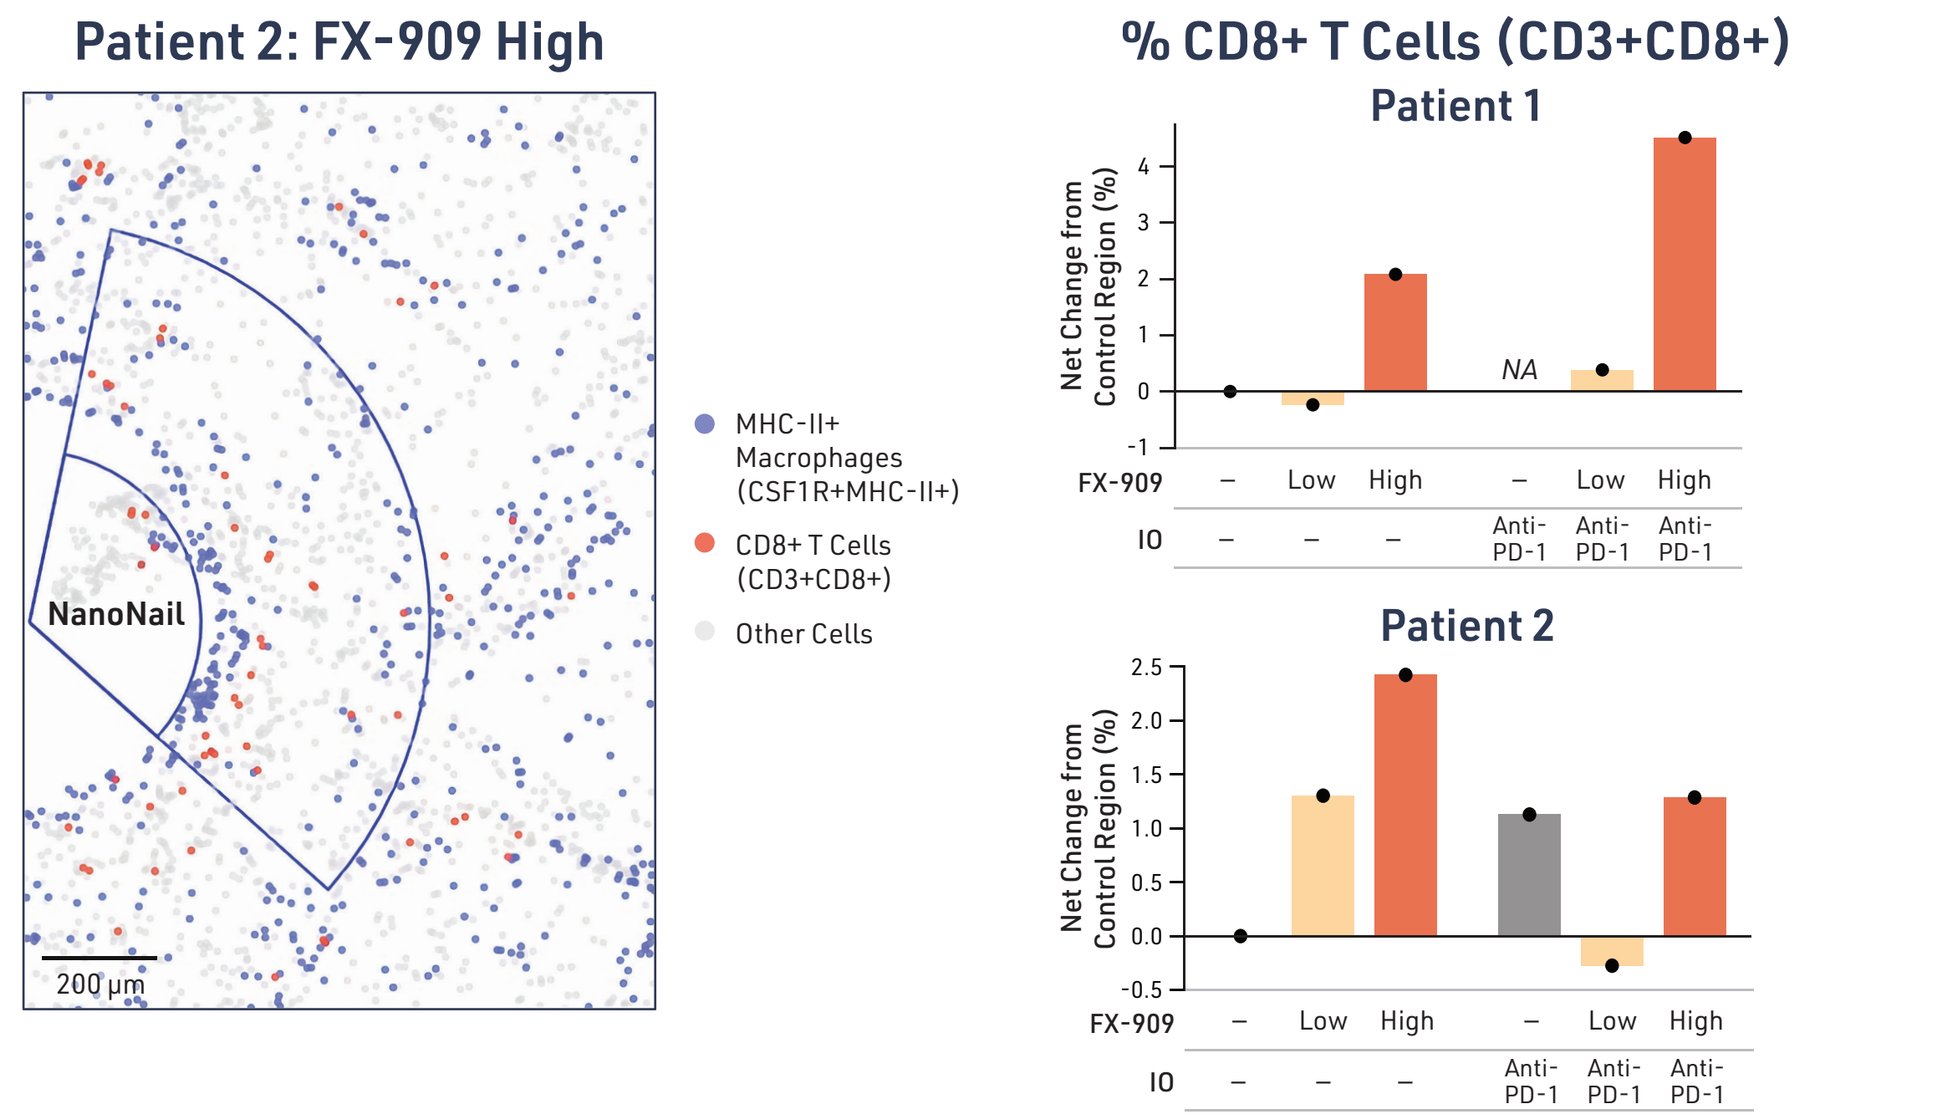

Patient 2: FX-909 High
NanoNail
200 µm
MHC-II+ Macrophages (CSF1R+MHC-II+)
CD8+ T Cells (CD3+CD8+)
Other Cells
% CD8+ T Cells (CD3+CD8+)
Patient 1
Patient 2
Net Change from Control Region (%)
NA
FX-909
IO
Low
High
Anti-PD-1

References **1.** Robertson et al, *Cell* 2017:171; **2.** Motley et al, *Eur J Cancer* 2022:174S1; **3.** Gao et al, AACR-NCI-EORTC 20250; **4.** Milowsky et al, *J Immunother Cancer* 2025:13(Suppl 3); **5.** Galsky et al, *Nat Med* 2026; **6.** Peruzzi et al, *Sci Transl Med* 2023:15; **7.** Jonas et al, *Sci Transl Med* 2015; **8.** Gjini et al, *J Clin Oncol* 2025; **9.** Muhlich et al, *Bioinformatics* 2022; **10.** Pachitariu, Rariden, & Stringer, *bioRxiv* 2025; **11.** Nirmal & Sorger, *J Open Source Softw* 2024.

Scan to View this Poster