GENOMIC PEROXISOME PROLIFERATOR-ACTIVATED RECEPTOR GAMMA (PPARG) STUDY: A SPONSORED TESTING PROGRAM TO IDENTIFY PATIENTS HARBORING GENETIC ALTERATIONS IN ADVANCED UC IN SUPPORT OF THE FX-909 PHASE 1 STUDY

Bijal Kakrecha, Adarsh Joshi, Kenneth Eagle, Rodney Marble, Michael L Meyers, Stefan Kirov, Evisa Gjini, Michaela Bowden
Flare Therapeutics, Cambridge, MA

Poster A024

INTRODUCTION

- The transcription factor PPARG is associated with the luminal lineage subtype representing ~65% of advanced urothelial cancer (UC) patients.
- Recurrent genetic alterations in PPARG including focal amplification, missense mutations, and fusions, as well as mutations in fibroblast growth factor receptor 3 (FGFR3) and hotspot mutations in PPARG's heterodimeric binding partner, retinoid X receptor alpha (RXRA), are characteristic of this luminal subtype.
- FX-909, a covalent PPARG inverse agonist, represents a promising first-in-class therapy for patients with advanced UC. FX-909-CLINPRO-1 [NCT05929235] is an open-label Phase 1 study to evaluate the safety, tolerability, pharmacokinetics, biologic activity, and preliminary clinical activity of FX-909. This study includes a Phase 1b expansion cohort, which will enroll advanced UC patients with genetic alterations in PPARG, RXRA, and/or FGFR3.
- Flare Therapeutics is sponsoring a testing program called Genomic PPARG Study (GPS) which allows investigators to offer the Tempus xT test to patients free of charge. GPS enables physicians to identify patients with these relevant genetic alterations in advance of the FX-909-CLINPRO-1 study.

METHODS

- Tumors are collected from patients with advanced or metastatic UC and sequenced using the Tempus xT assay (648 genes DNAseq panel at 500x coverage), and if sufficient tissue is provided, the Tempus xR assay (whole transcriptome RNAseq). A clinical report is generated and returned to the physician to guide selection of approved therapies, and if appropriate, to support eligibility for inclusion in the study. FlareTx is provided with de-identified genomic data, where assessment of the prevalence of genetic alternations such as copy number variation (CNV), fusions and mutations (single nucleotide variants) in PPARG, RXRA and FGFR3 are undertaken. Additionally, molecular classification as luminal (luminal papillary, luminal, and luminal infiltrated subtypes) or non-luminal (basal-squamous and neuronal subtypes) is performed using non-negative matrix factorization (NMF) rank 5 following the Robertson method[1], excluding PPARG from the gene set to eliminate the inference of bias. PPARG mean expression value are normalized by transcripts-per-million (TPM) and reported as Log2[TPM+1]. The data represented here is an early snapshot of the first 25 patients screened through the GPS sponsored testing program.

PROGRAM ELIGIBILITY CRITERIA

- An individual must be 18 years or older.
- The submitted specimen has not been previously tested under the Program.
- The individual has been diagnosed with advanced (unresectable) or metastatic urothelial cancer requiring systemic therapy.
 - Patient who have progressed through at least one line of systemic therapy in the advanced and metastatic setting

 or
 - Patient newly diagnosed with advanced and metastatic urothelial cancer but have not yet received systemic therapy in the advanced and metastatic setting.

FIGURE 1. PPARG Maintains Luminal Identity in Urothelial Cancer[3]

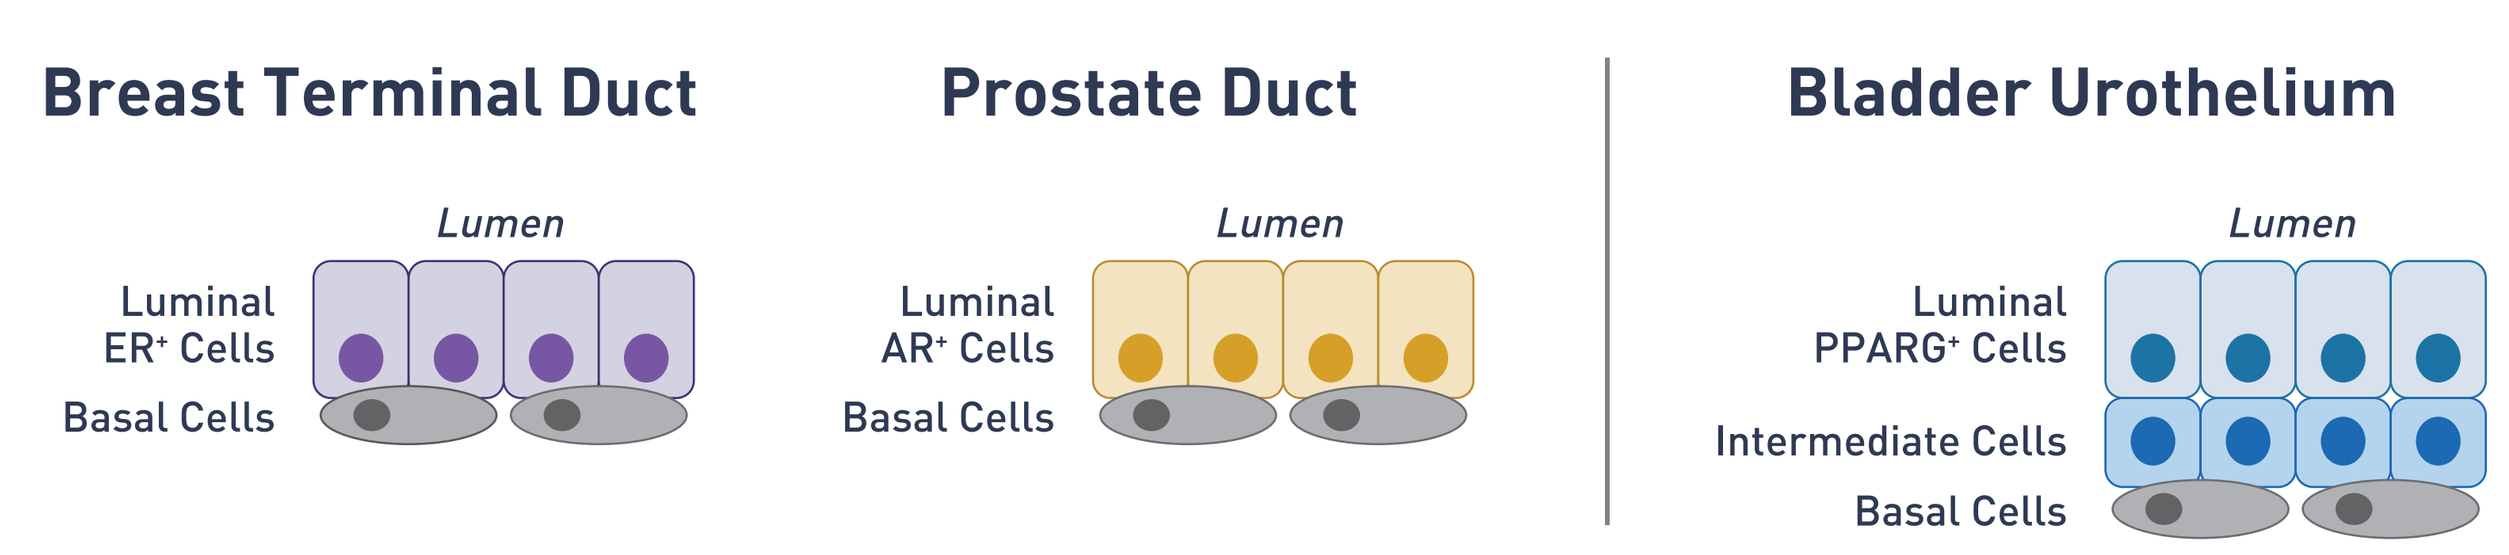
Breast Terminal Duct
Lumen
Luminal ER+ Cells
Basal Cells
Prostate Duct
Lumen
Luminal AR+ Cells
Basal Cells
Bladder Urothelium
Lumen
Luminal PPARG+ Cells
Intermediate Cells
Basal Cells

- Targeting cell identity is a clinically validated therapeutic strategy exemplified with ER and AR approved agents in breast and prostate cancer, respectively.

FIGURE 3. The **G**enome **P**PARG **S**tudy or **GPS*** is a Sponsored Testing Program that Runs Concurrently with the FX-909 Clinical Study Priming the Pump for Enrollment

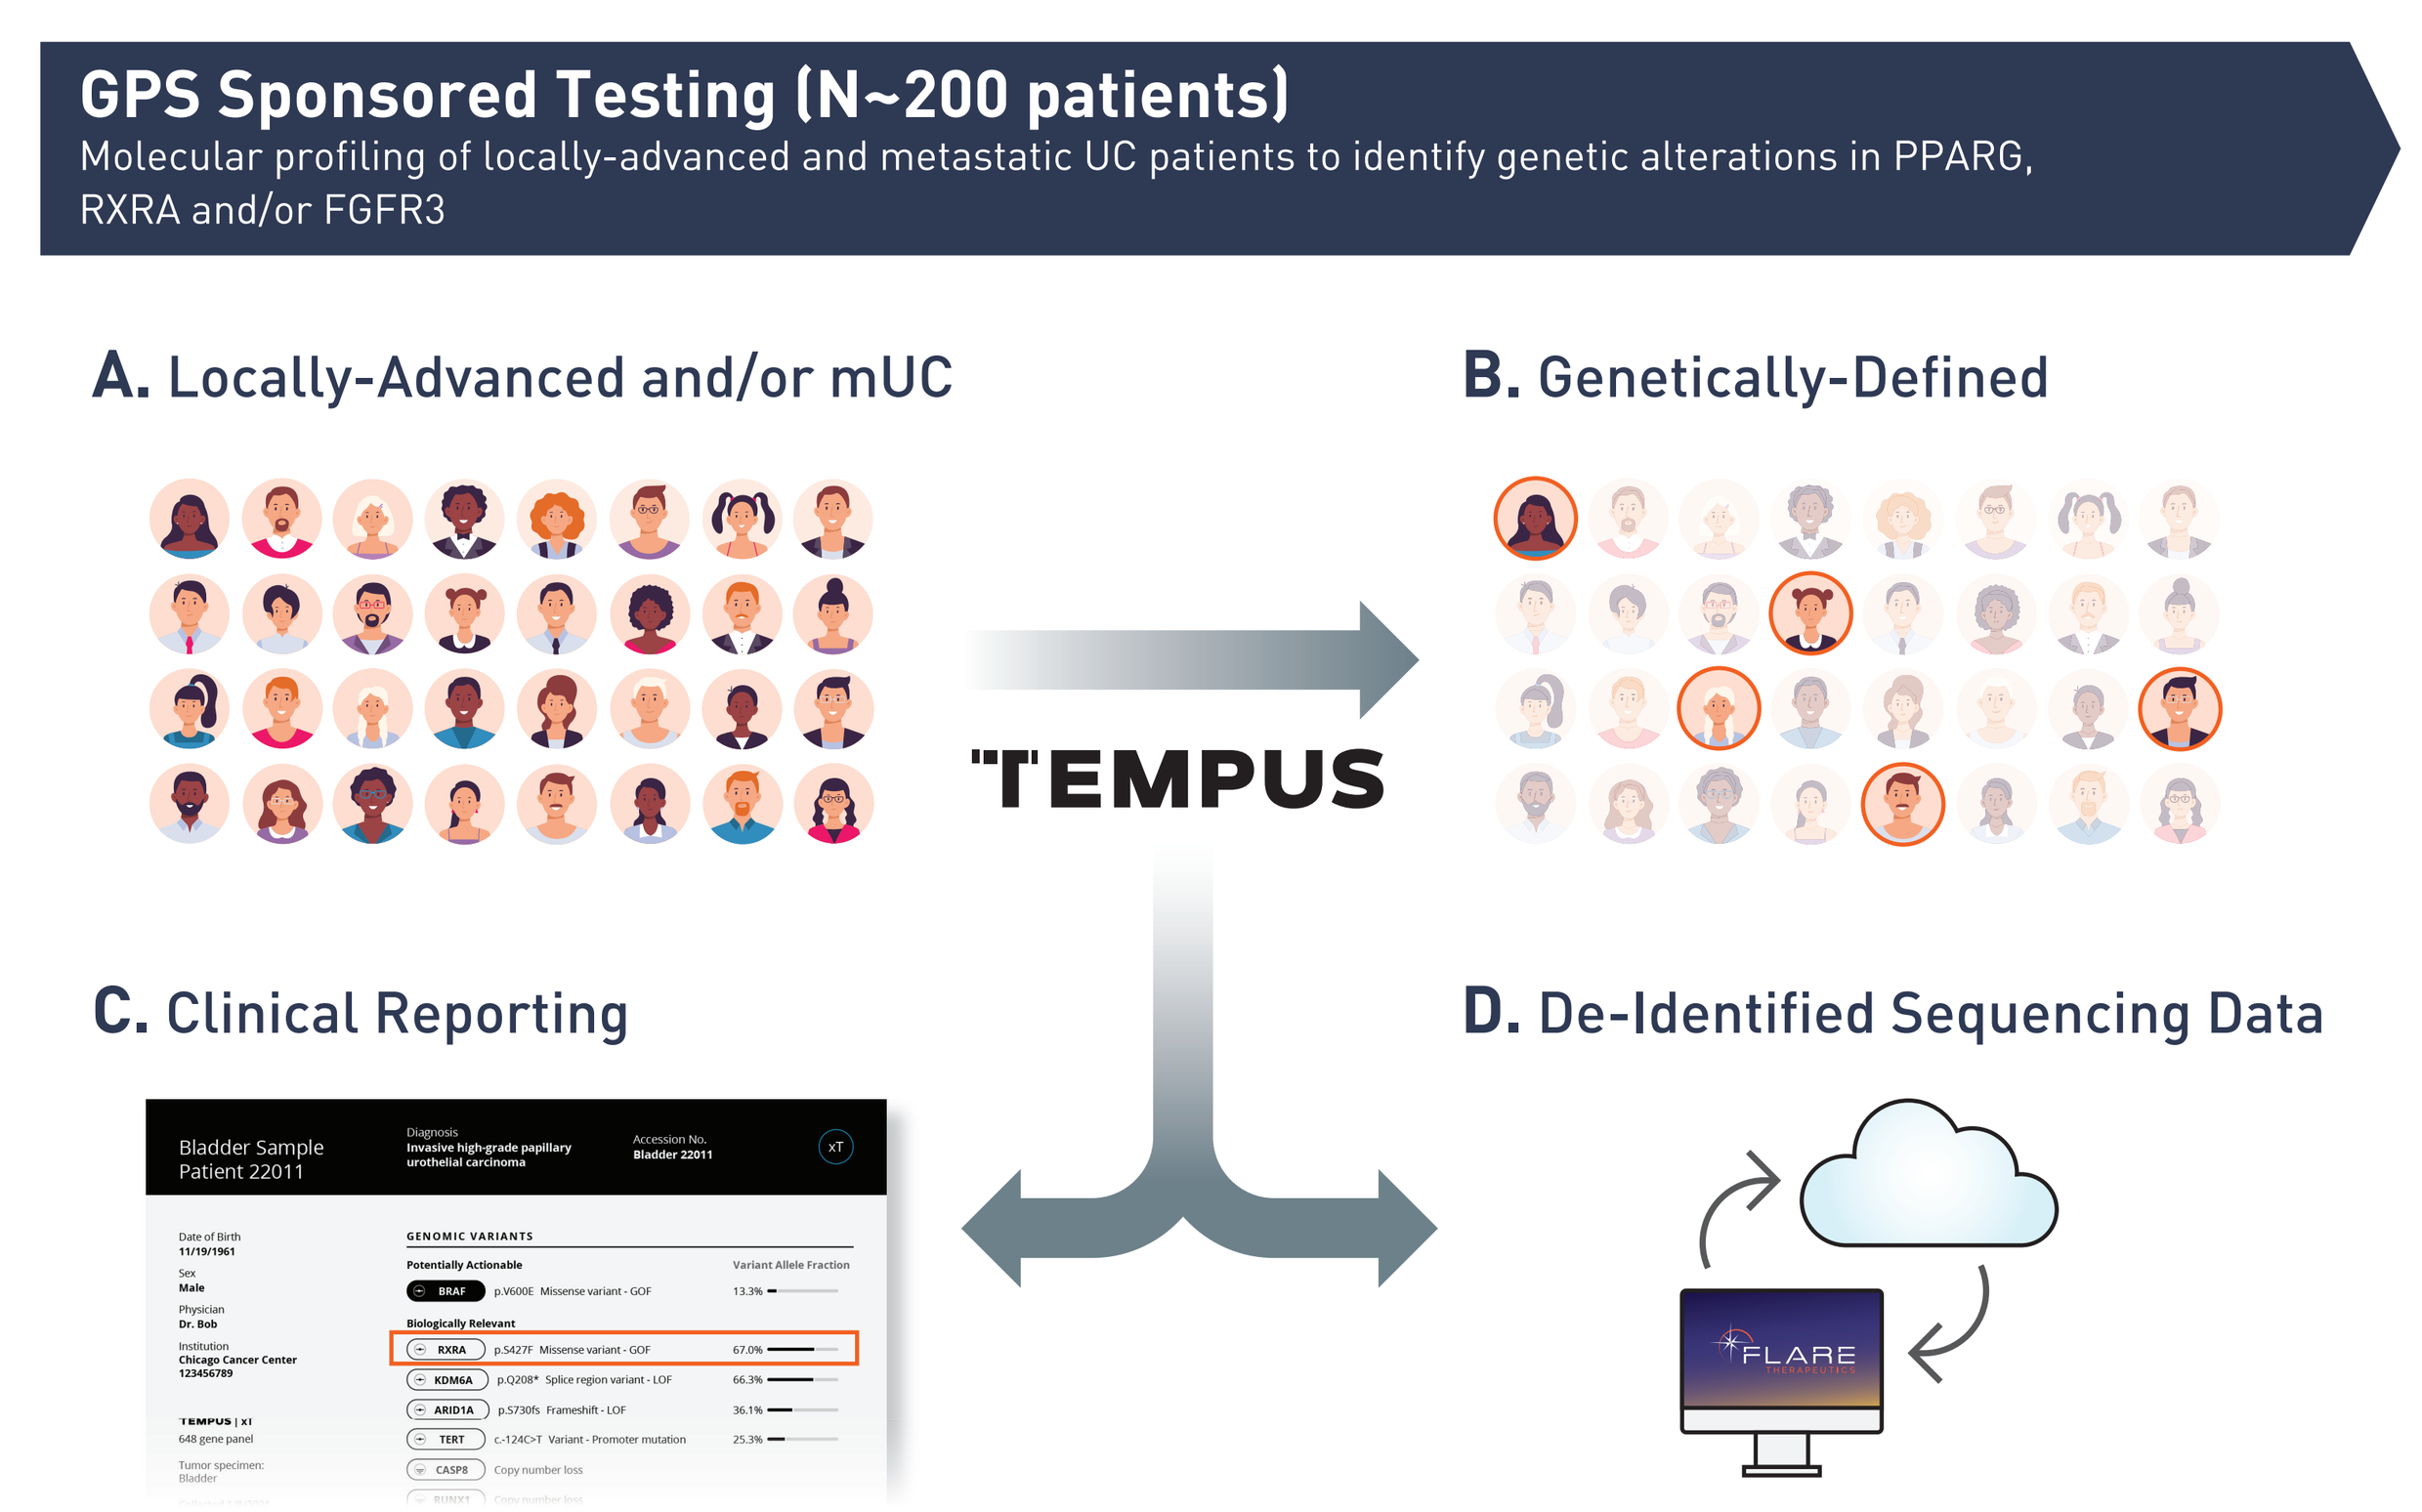
GPS Sponsored Testing (N~200 patients)
Molecular profiling of locally-advanced and metastatic UC patients to identify genetic alterations in PPARG, RXRA and/or FGFR3
A. Locally-Advanced and/or mUC
B. Genetically-Defined
TEMPUS
C. Clinical Reporting
D. De-Identified Sequencing Data
FLARE

*GPS as in navigation tool to enroll genetically-defined patients to Phase 1 protocol.

A. GPS is open to all advanced UC patient who require systemic therapy
B. Tempus provides commercial CAP/CLIA molecular profiling (xT, xR test)
C. Physicians receive a clinical report to aid treatment decision-making for patients
D. FlareTx gathers real-time, real-world translational data

FIGURE 2. PPARG Status Defines the Luminal Lineage in Advanced UC Patients[1-2]

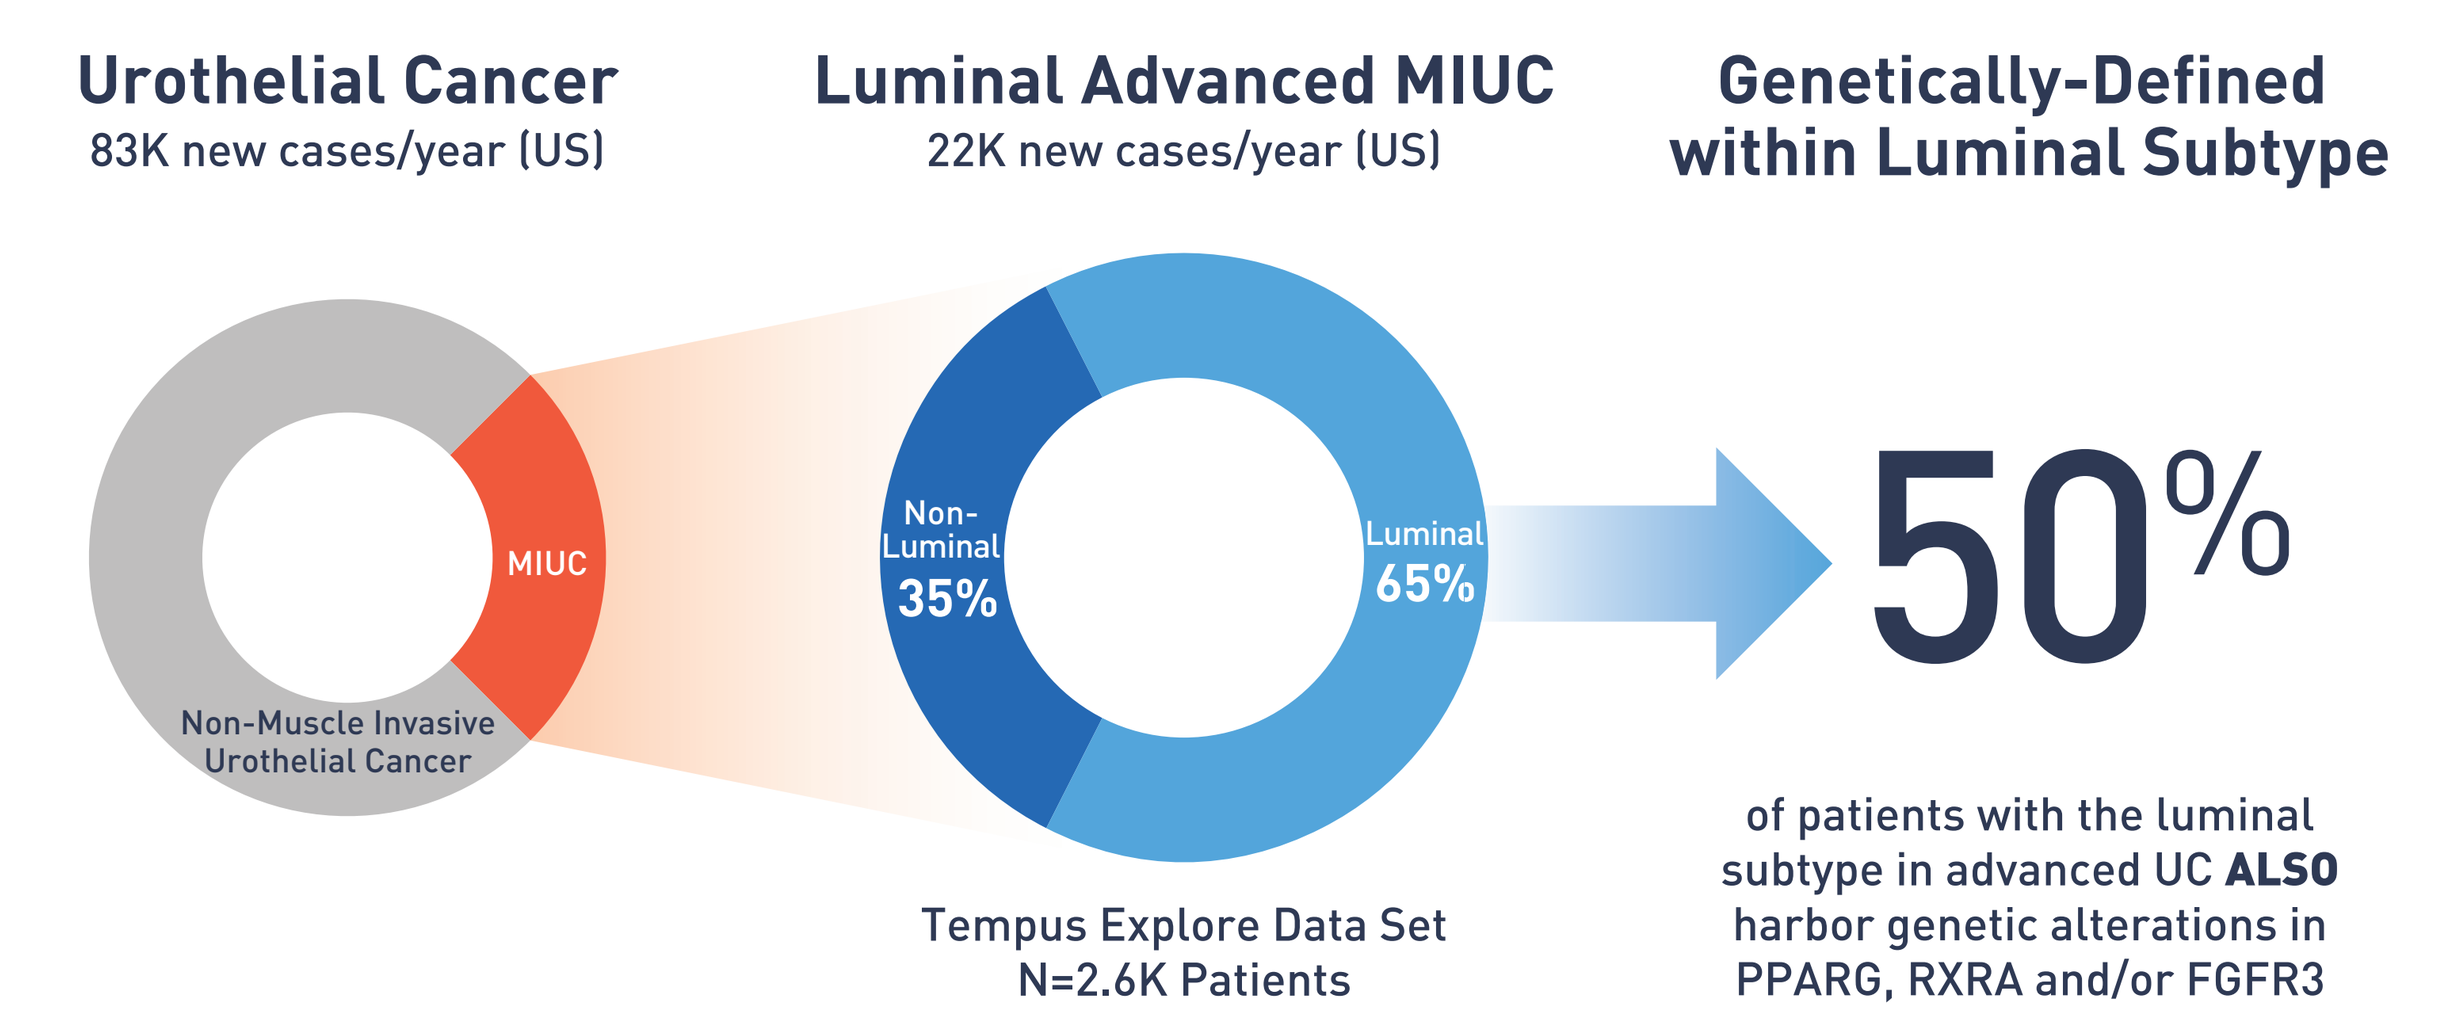
Urothelial Cancer
83K new cases/year (US)
Luminal Advanced MIUC
22K new cases/year (US)
Genetically-Defined within Luminal Subtype
MIUC
Non-Muscle Invasive Urothelial Cancer
Non-Luminal 35%
Luminal 65%
Tempus Explore Data Set
N=2.6K Patients
50%
of patients with the luminal subtype in advanced UC ALSO harbor genetic alterations in PPARG, RXRA and/or FGFR3

- Exploration of molecular real-world data (MRWD) set of advanced MIUC patients revealed that 65% of these tumors are classified as having a luminal subtype, with 50% of luminal cases harboring genetic alterations in PPARG, RXRA, and/or FGFR3.

FIGURE 4. GPS Sponsored Testing Program Operating Model

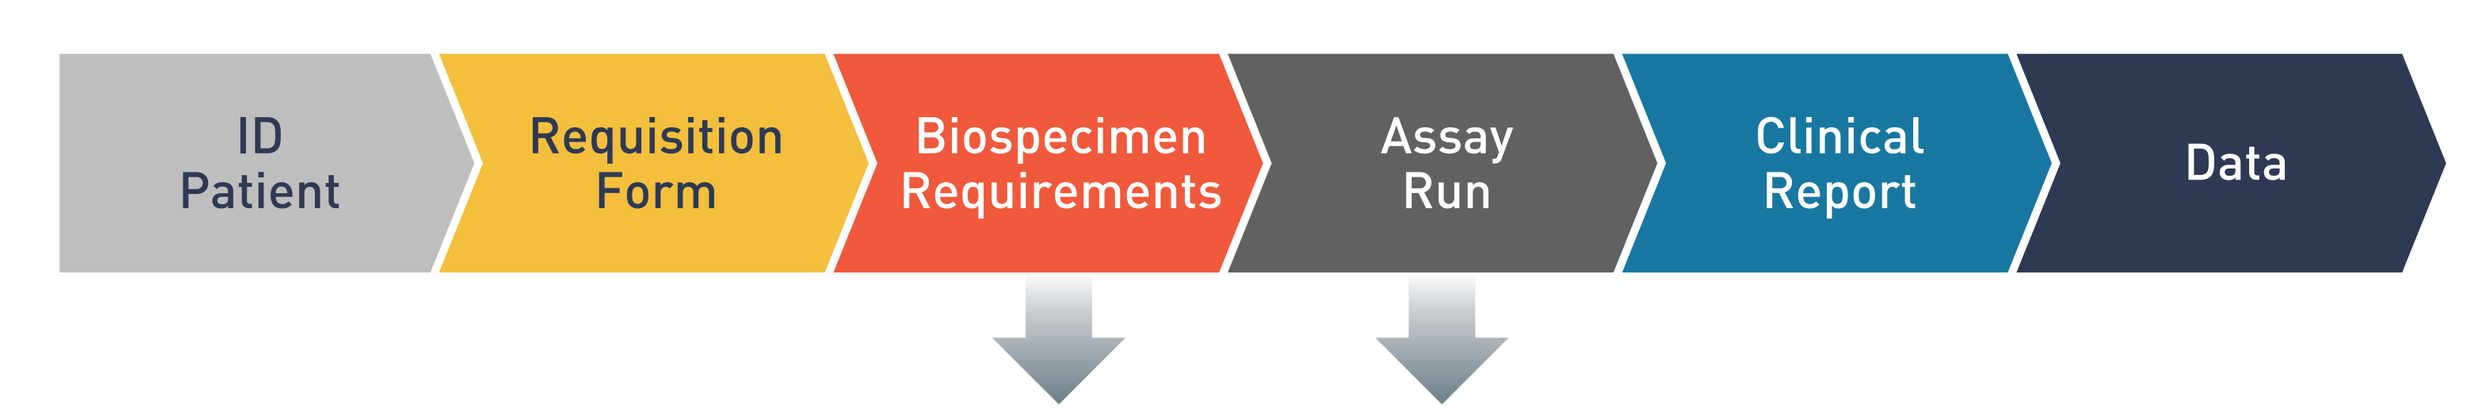
ID Patient
Requisition Form
Biospecimen Requirements
Assay Run
Clinical Report
Data

Biospecimen Requirements	
Tumor Requirement	At least 20% of the sample by ratio of tumor nuclei to benign nuclei
Solid Tumor	1 FFPE block ***or***
	10 x 5µm unstained slides + 1 H&E ***or***
	Minimum 210ng yield, ≥2ng/µl concentration
Matched Normal	2 Streck tubes of 8.5 ml of blood ***or***
	Saliva

Assay Run	
Targeted DNAseq Panel & Whole Transcriptome RNAseq Panel	648 gene tumor/normal matched, including PPARG, RXRA, & FGFR3
Output	DNA (Tumor) 500x coverage
	RNA 50M reads depth
Platform	Illumina NextSeq™ 500

- After identifying eligible patients, sites work directly with Tempus to order the xT test using a program-specific Flare Therapeutics Sponsored Testing Program Requisition Form. Physicians receive a clinical report and FlareTx receives de-identified raw and processed data to evaluate the prevalence of genetic alterations.

FIGURE 5. Early Evidence from Real-Time GPS Testing Correlates with Large Scale Molecular Real-World Data[1-2]

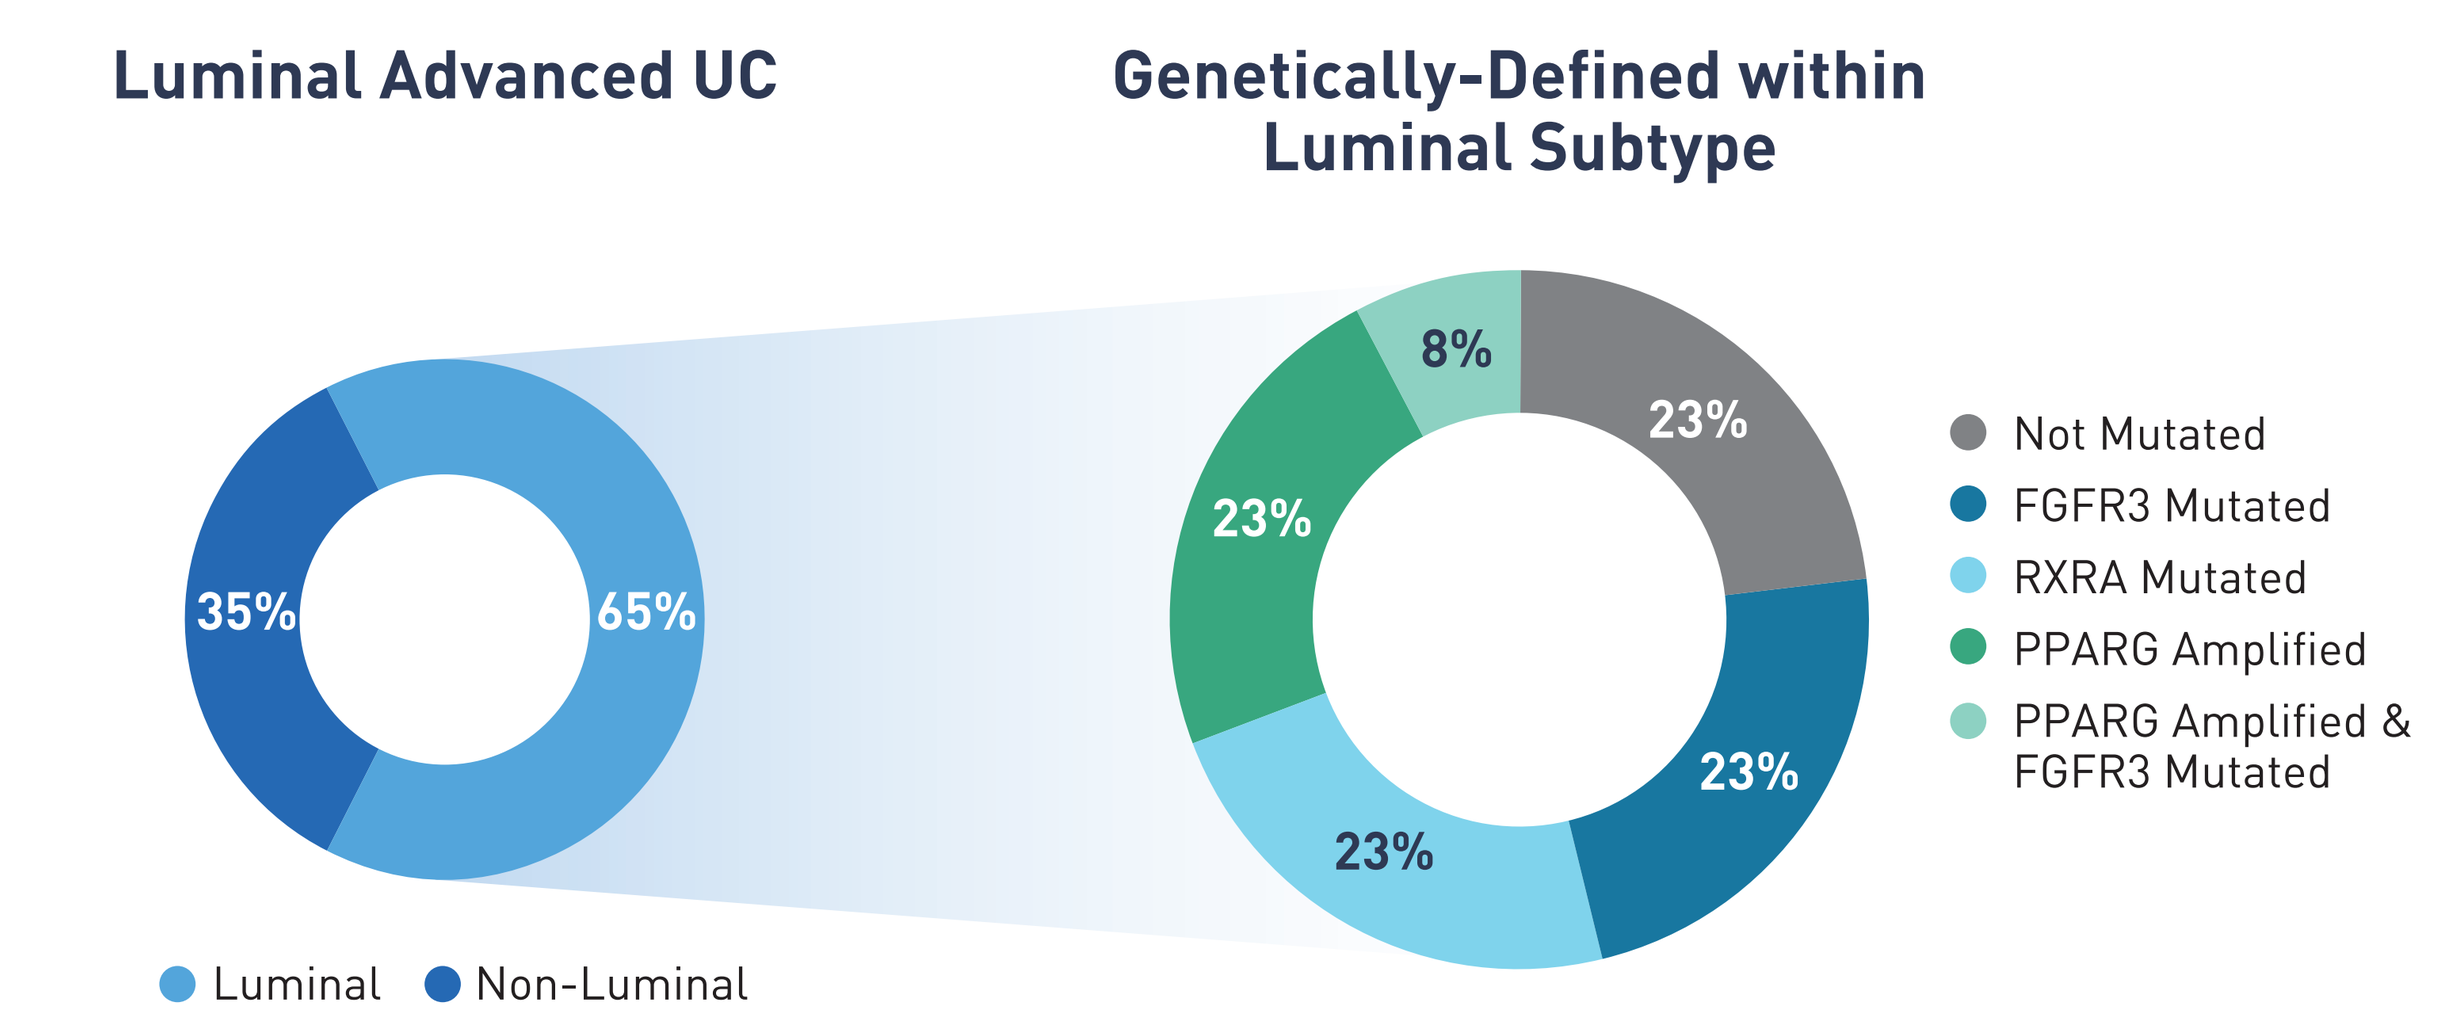
Luminal Advanced UC
Genetically-Defined within Luminal Subtype
35%
65%
Luminal
Non-Luminal
8%
23%
23%
23%
23%
Not Mutated
FGFR3 Mutated
RXRA Mutated
PPARG Amplified
PPARG Amplified & FGFR3 Mutated

- Consistent with the TCGA subtype distribution, 65% of patients were luminal and 35% were non-luminal (basal-squamous and neuronal). As of data cut in February 2024, of the patients with luminal subtype >50% harbor one or more genetic alteration in PPARG, RXRA, and/or FGFR3.

TABLE 1. Preliminary Analysis of GPS PPARG mRNA Expression Correlates with Large Molecular Real-World Data[2]

Molecular Subtypes	Average PPARG mRNA Expression Level Log2[TPM+1] [95% CI]	
	Tempus MRWD	GPS Sponsored Testing Program
Luminal Lineage	7.78 [7.71-7.85]	7.90 [7.44-8.36]
Non-Luminal Lineage	5.47 [5.34-5.61]	4.67 [3.55-5.79]

Abbreviations: **CI** – Confidence interval; **TPM** – Transcripts per million

- Consistent with the large-scale Tempus real-world study previously published, PPARG mRNA expression is significantly higher in luminal lineage compared to non-luminal linage (Wilcoxon, p=0.00042).

FIGURE 6. FX-909-CLINPRO-1 Phase 1 Study Design [NCT05929235]

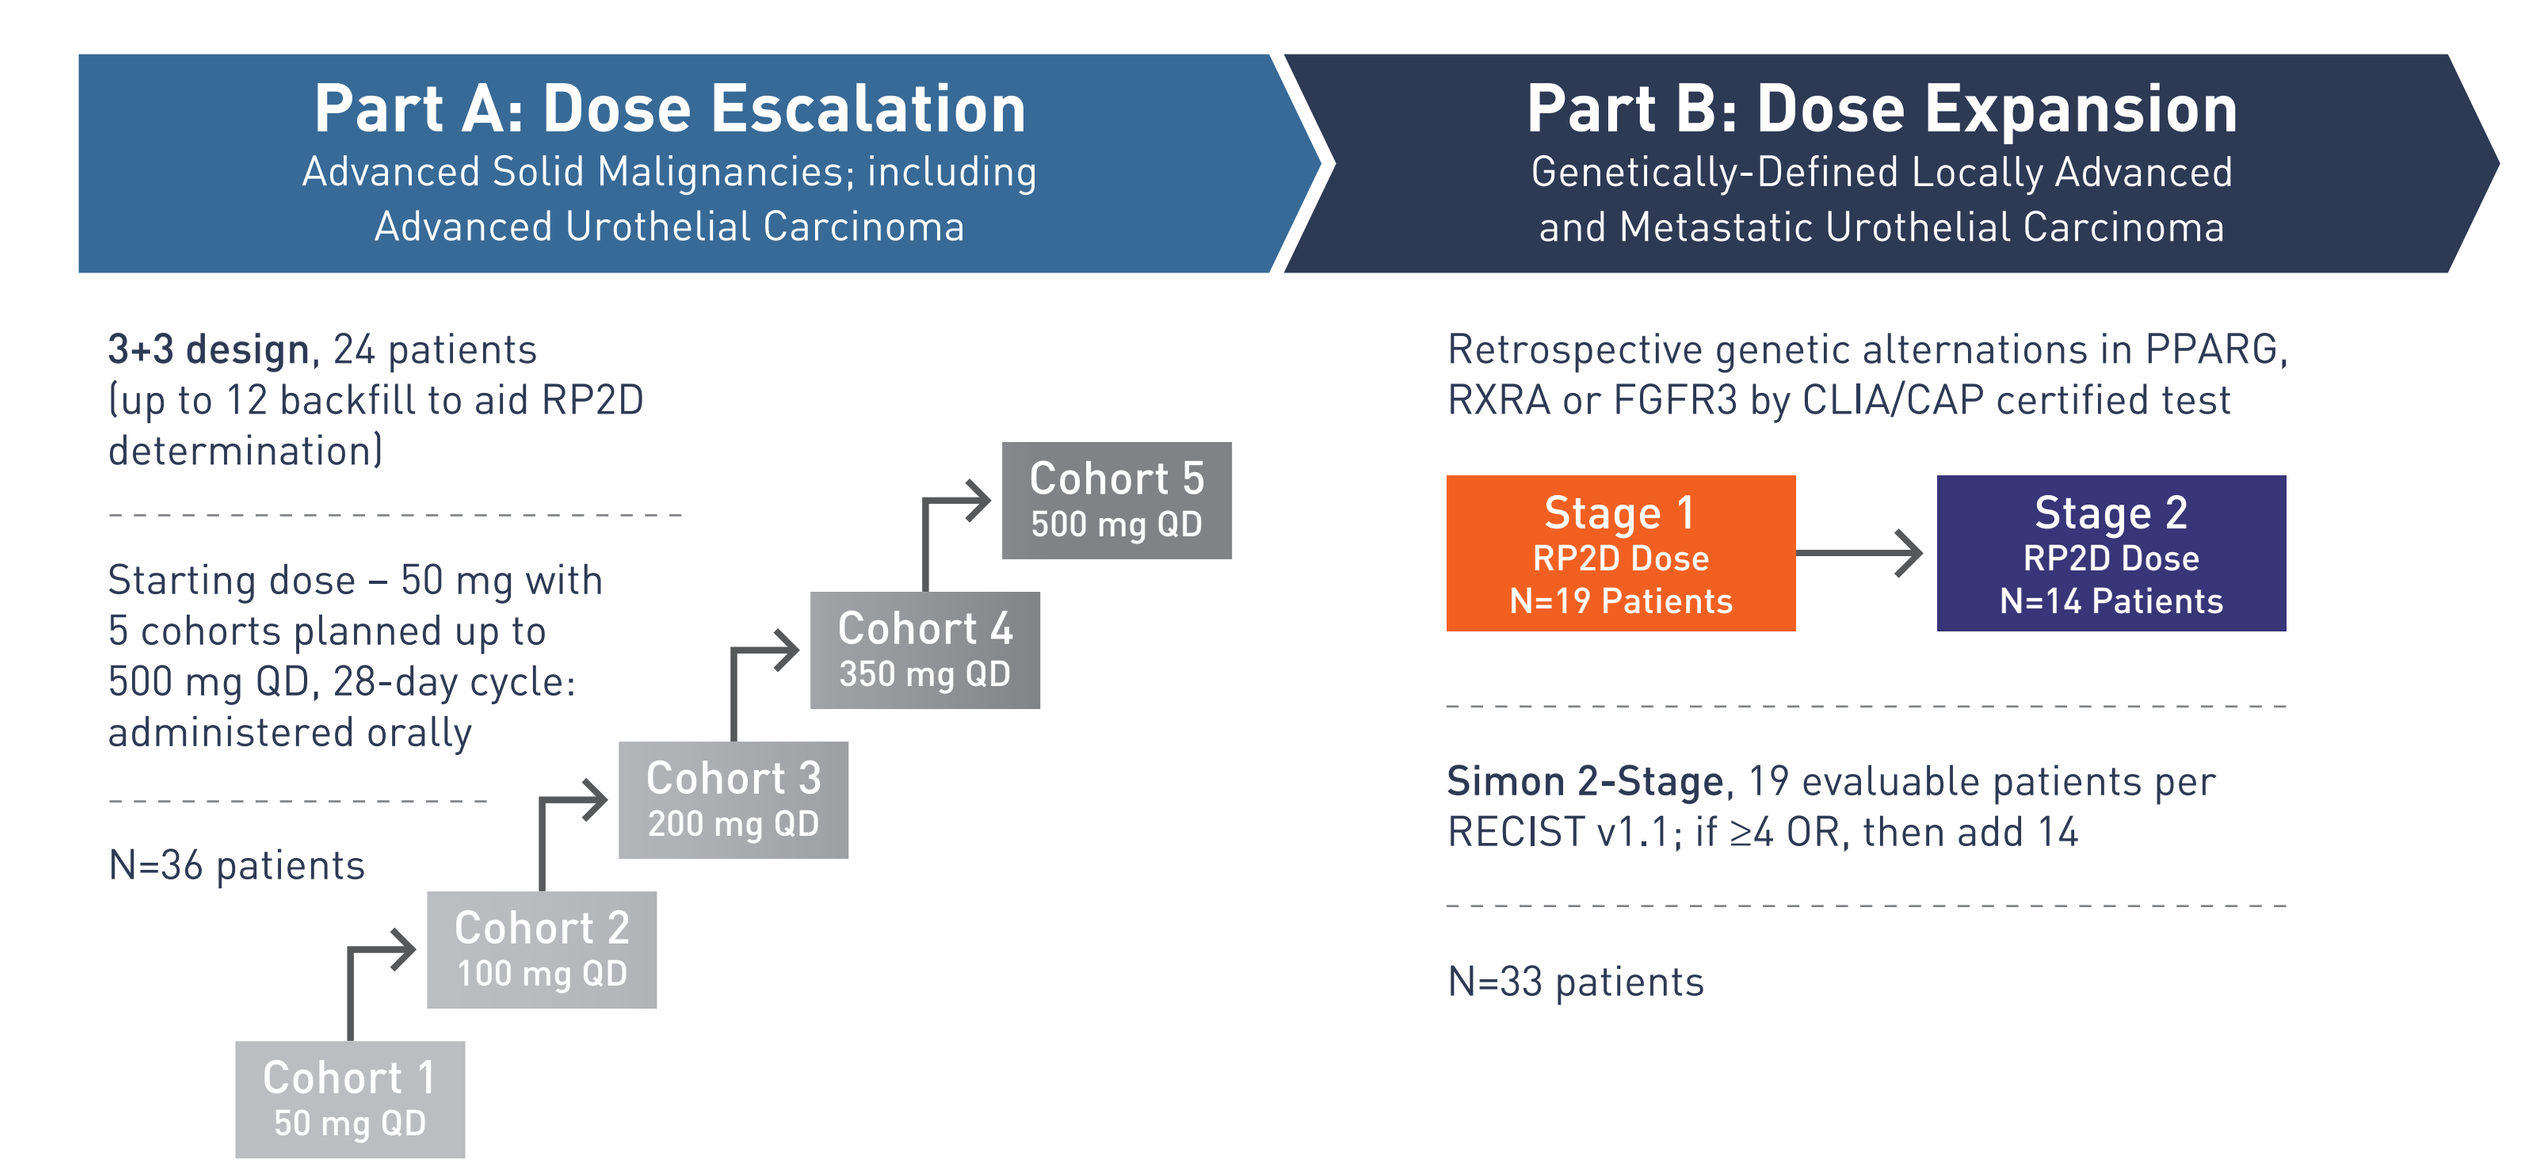
Part A: Dose Escalation
Advanced Solid Malignancies; including Advanced Urothelial Carcinoma
Part B: Dose Expansion
Genetically-Defined Locally Advanced and Metastatic Urothelial Carcinoma
3+3 design, 24 patients (up to 12 backfill to aid RP2D determination)
Starting dose – 50 mg with 5 cohorts planned up to 500 mg QD, 28-day cycle: administered orally
N=36 patients
Cohort 1 50 mg QD
Cohort 2 100 mg QD
Cohort 3 200 mg QD
Cohort 4 350 mg QD
Cohort 5 500 mg QD
Retrospective genetic alternations in PPARG, RXRA or FGFR3 by CLIA/CAP certified test
Stage 1 RP2D Dose N=19 Patients
Stage 2 RP2D Dose N=14 Patients
Simon 2-Stage, 19 evaluable patients per RECIST v1.1; if ≥4 OR, then add 14
N=33 patients

Abbreviations: **RP2D** – Recommended Phase 2 dose; **QD** – Once daily; **OR** – Overall response

SUMMARY

- The GPS program is open at two active Phase 1 sites for the FX-909-CLINPRO-1 [NCT05929235] study, with more planned as the study advances towards Phase 1B dose expansion.
- The sponsored testing program runs concurrently with the FX-909 study and is a novel precision strategy to maximize identification of advanced UC patients with actionable genetic alterations to potentially enroll in the clinical study.
- Since opening the GPS program, Tempus has validated PPARG amplification (CN>8) as a clinical reportable across all xT testing.
- Precision oncology approaches such as the GPS sponsored testing program accelerate our understanding of the underlying MOA from the patient perspective, allowing us to anticipate the right patients most likely to benefit from treatment with FX-909, a PPARG inverse agonist, as well as offering new insights to physicians and their patients as they decide on therapeutic options in a dynamic clinical landscape.

Scan to Visit the ClinicalTrial.gov FX-909-CLINPRO-1 Study Page

Scan to View Flare Publications

References: **1.** Robertson AG et al. (2017) *Cell* 171:540-556; **2.** Motley W et al. 2022 EORTC-NCI-AACR, Abstract 333; **3.** Iyer G et al. 2024 ASCO GU, Abstract TPS709.