A PHASE 1, FIRST-IN-HUMAN, DOSE-ESCALATION AND EXPANSION STUDY OF FX-909 IN PATIENTS WITH ADVANCED SOLID MALIGNANCIES, INCLUDING ADVANCED UROTHELIAL CARCINOMA

FLARE THERAPEUTICS

Gopa Iyer[1], Xin Gao[2], Drew Rasco[3], Matthew Milowsky[4], Benjamin Garmezy[5], I. Ismael Rodriguez Rivera[6], Jennifer Tepper[7], Melissa Moles[7], Evisa Gjini[7], Michaela Bowden[7], Michael L Meyers[7], Joaquin Bellmunt Molins[8], Matthew Galsky[9]

[1]Memorial Sloan Kettering Cancer Center, New York, NY; [2]Massachusetts General Hospital, Boston, MA; [3]START San Antonio, San Antonio, TX; [4]University of North Carolina, Lineberger Comprehensive Cancer Center, Chapel Hill, NC; [5]Sarah Cannon Research Institute, Nashville, TN; [6]NEXT San Antonio, San Antonio, TX; [7]Flare Therapeutics, Inc, Cambridge, MA; [8]Dana-Farber Cancer Institute, Boston, MA; [9]Mount Sinai Health System, New York, NY

TPS709

INTRODUCTION AND STUDY RATIONALE

- Urothelial carcinoma (UC) remains a disease with high unmet need, in which the 5-year survival for patients with locally advanced muscle-invasive disease is < 40% in the USA and falls to 8% in patients with distant metastases.[1-3]
- Novel agents are needed to improve outcomes in UC treated with standard of care (eg, platinum-doublet chemotherapy, PD-(L)1 checkpoint inhibition, and antibody-drug conjugates (enfortumab vedotin and sacituzumab govitecan).
- Peroxisome proliferator-activated receptor gamma (PPARG) plays an essential lineage-determining role in normal urothelium[4,5] and is crucial for urothelial homeostasis and regeneration. Activation of PPARG-dependent transcription, either owing to mutations in its binding partner RXRA or amplification of the *PPARG* gene, occurs in 30% of luminal tumors.[6-9] Mutations in FGFR3 are more frequently observed in cancers with high expression of PPARG that are classified as luminal.
- FX-909 is a new, orally available molecular entity that irreversibly, specifically, and potently inhibits basal- and ligand-activated transcription by PPARG. FX-909 is being investigated in the clinical trial FX-909-CLINPRO-1 (NCT05929235).

DISEASE BACKGROUND

PPARG maintains luminal identity in UC

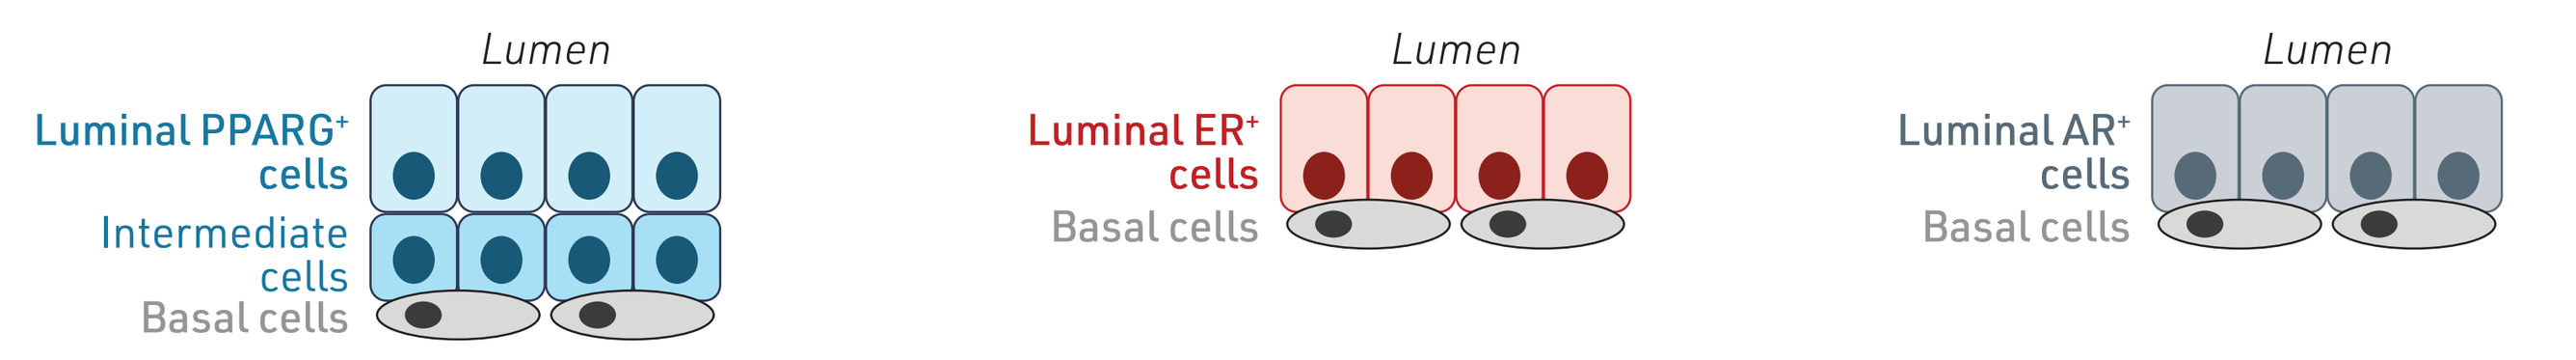
Bladder Urothelium
Breast Terminal Duct
Prostate Duct
Lumen
Luminal PPARG+ cells
Intermediate cells
Basal cells
Luminal ER+ cells
Basal cells
Luminal AR+ cells
Basal cells

Urothelial Cancer Cell of Origin
- 50-60% luminal
- PPARG maintains luminal identity
- PPARG-RXRA mutations occur in 30% of cases with advanced MIUC

Breast Cancer Cell of Origin
- 60-70% luminal
- ER maintains luminal identity
- ER inhibitors are backbone therapies in luminal breast cancer

Prostate Cancer Cell of Origin
- 100% luminal
- AR maintains luminal identity
- AR inhibitors are backbone therapies in prostate cancer

PPARG Genetic Status Defines the Luminal Lineage in a Large Molecular Real-World Cohort of Advanced UC[1,8-10]

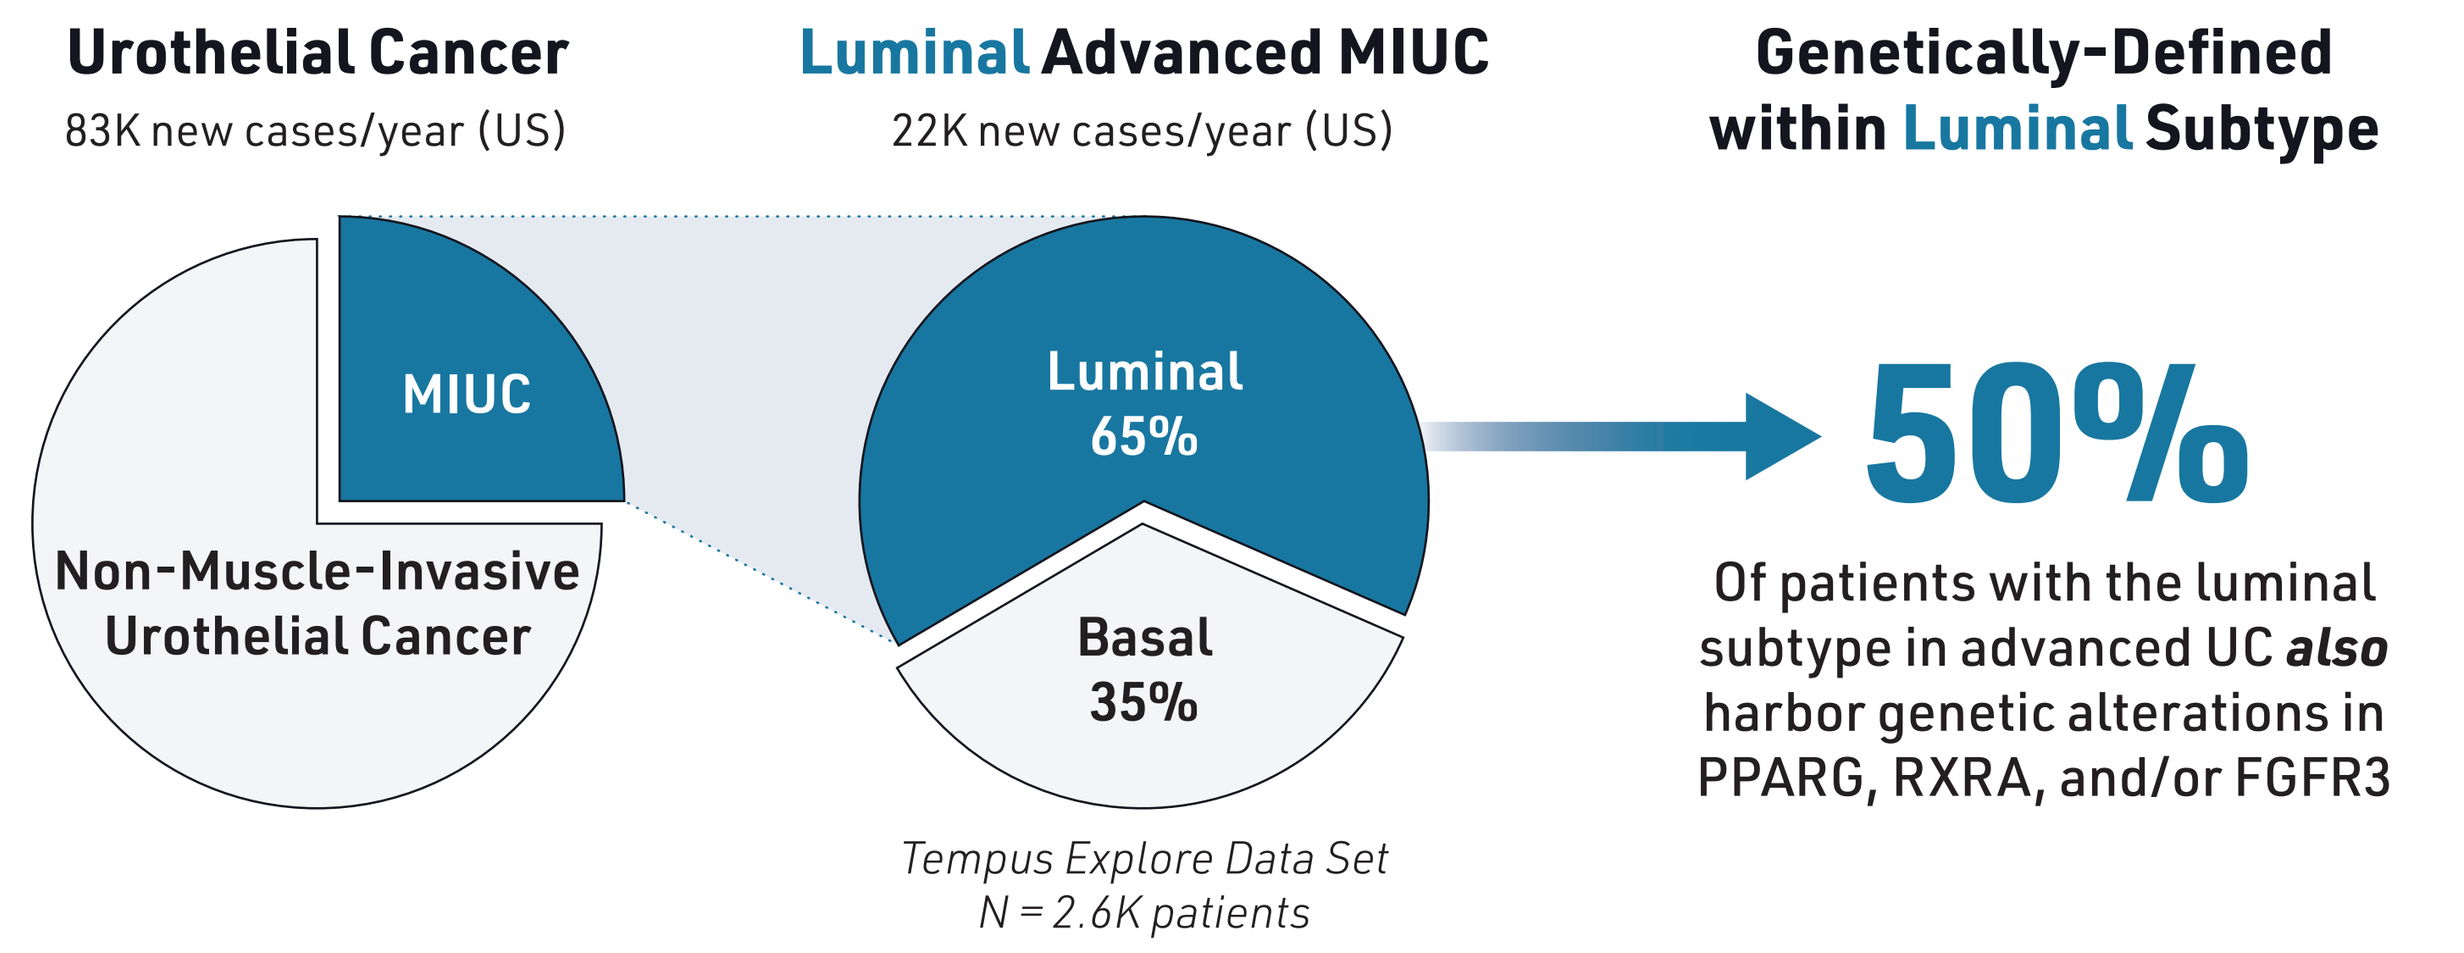
Urothelial Cancer
83K new cases/year (US)
Luminal Advanced MIUC
22K new cases/year (US)
Genetically-Defined within Luminal Subtype
MIUC
Non-Muscle-Invasive Urothelial Cancer
Luminal 65%
Basal 35%
50%
Of patients with the luminal subtype in advanced UC *also* harbor genetic alterations in PPARG, RXRA, and/or FGFR3
Tempus Explore Data Set
N = 2.6K patients

FX-909-CLINPRO-1 PHASE 1 STUDY

Study Design

Evaluation of the safety and tolerability, PK, PD, and preliminary clinical activity of FX-909 given orally in patients with advanced solid malignancies.

PART A: DOSE ESCALATION (N ≤ 36)
Advanced Solid Malignancies, Including Advanced UC

3+3 design, 24 patients (up to 12 backfill to aid RP2D determination)

Cohort 1 50 mg → Cohort 2 100 mg → Cohort 3 200 mg → Cohort 4 350 mg → Cohort 5 500 mg

28-day cycle, dose administered PO, QD

PART B: DOSE EXPANSION (N = 33)
Genetically Defined*, Measurable Locally Advanced and Metastatic UC

Simon 2-Stage, 19 evaluable patients per RECIST v1.1; if ≥ 4 OR, then add 14

Stage 1 RP2D dose N = 19 → Stage 2 RP2D dose N = 14

Genetic alterations in PPARG, RXRA, or FGFR3 by CLIA/CAP-certified test

Objectives and Endpoints

	Objectives	Endpoints
Primary (Pt. A)	To assess safety and tolerability	Incidence of DLTs, SAE/AEs
Primary (Pt. B)	To evaluate antitumor activity in locally advanced (unresectable) and metastatic UC	OR
Secondary	To determine RP2D, assess PK and anti-tumor activity	Plasma and urine PK, ORR, DOR, DCR, PFS, and OS per RECIST version 1.1 criteria
Exploratory	To evaluate PD, exposure-response, exposure-safety, patient selection biomarkers, immunomodulatory effect	Skin & periphery PD (PPARG target genes), ctDNA, PPARG IHC, WES, RNAseq, PD-L1 IHC, CD8 IHC, CD8 topology, and scRNAseq on PBMC

Patient Screening Criteria

Inclusion Criteria
Age ≥ 18 years
ECOG Score 0-2
Screening Laboratory Values Meet the criteria outlined per protocol, including: » HbA1c < 7.0% » Fasting glucose ≤ 140 mg/dL » Fasting triglycerides ≤ 200 mg/dL » CrCl > 60 mL/min
Part A (Dose Escalation) » Any advanced solid cancer » Measurable disease **NOT** required
Part B (Dose Expansion) » Advanced UC » Measurable Disease per RECIST v1.1 » Mutation/Amplification/Fusion in PPARG, RXRA, or FGFR3

Exclusion Criteria
T1DM or inadequately controlled T2DM **(Patients on insulin are excluded)**
QTcF > 470 msec in screening
Active diagnosis of lipodystrophy/lipoatrophy
Uncontrolled or symptomatic CNS metastases
Chronic or recurrent pancreatitis at any time, or diagnosis of acute pancreatitis < 6 mos of Screening

Exploratory Biomarkers and Collection Timepoints

Assessment	Biospecimen	Screening	C1D1	C1D15	C2D1	C3D1	C4D1	EOT
PK	Plasma	●	●	●	●	●	●	●
PK	Urine	●	●	●				
PD	Skin biopsy	●		●				
PD	Plasma	●	●	●	●	●	●	●
Patient Selection Biomarker Analysis	FFPE (IHC, H&E)	●						
Patient Selection Biomarker Analysis	FFPE (WES, RNAseq)*	●				optional ○		optional ○
Patient Selection Biomarker Analysis	ctDNA	●			●	●	●	●
Immunoprofiling	PBMC (scRNAseq)	●			●	●	●	●

*supported by ctDNA analysis

Phase 1 US Sites

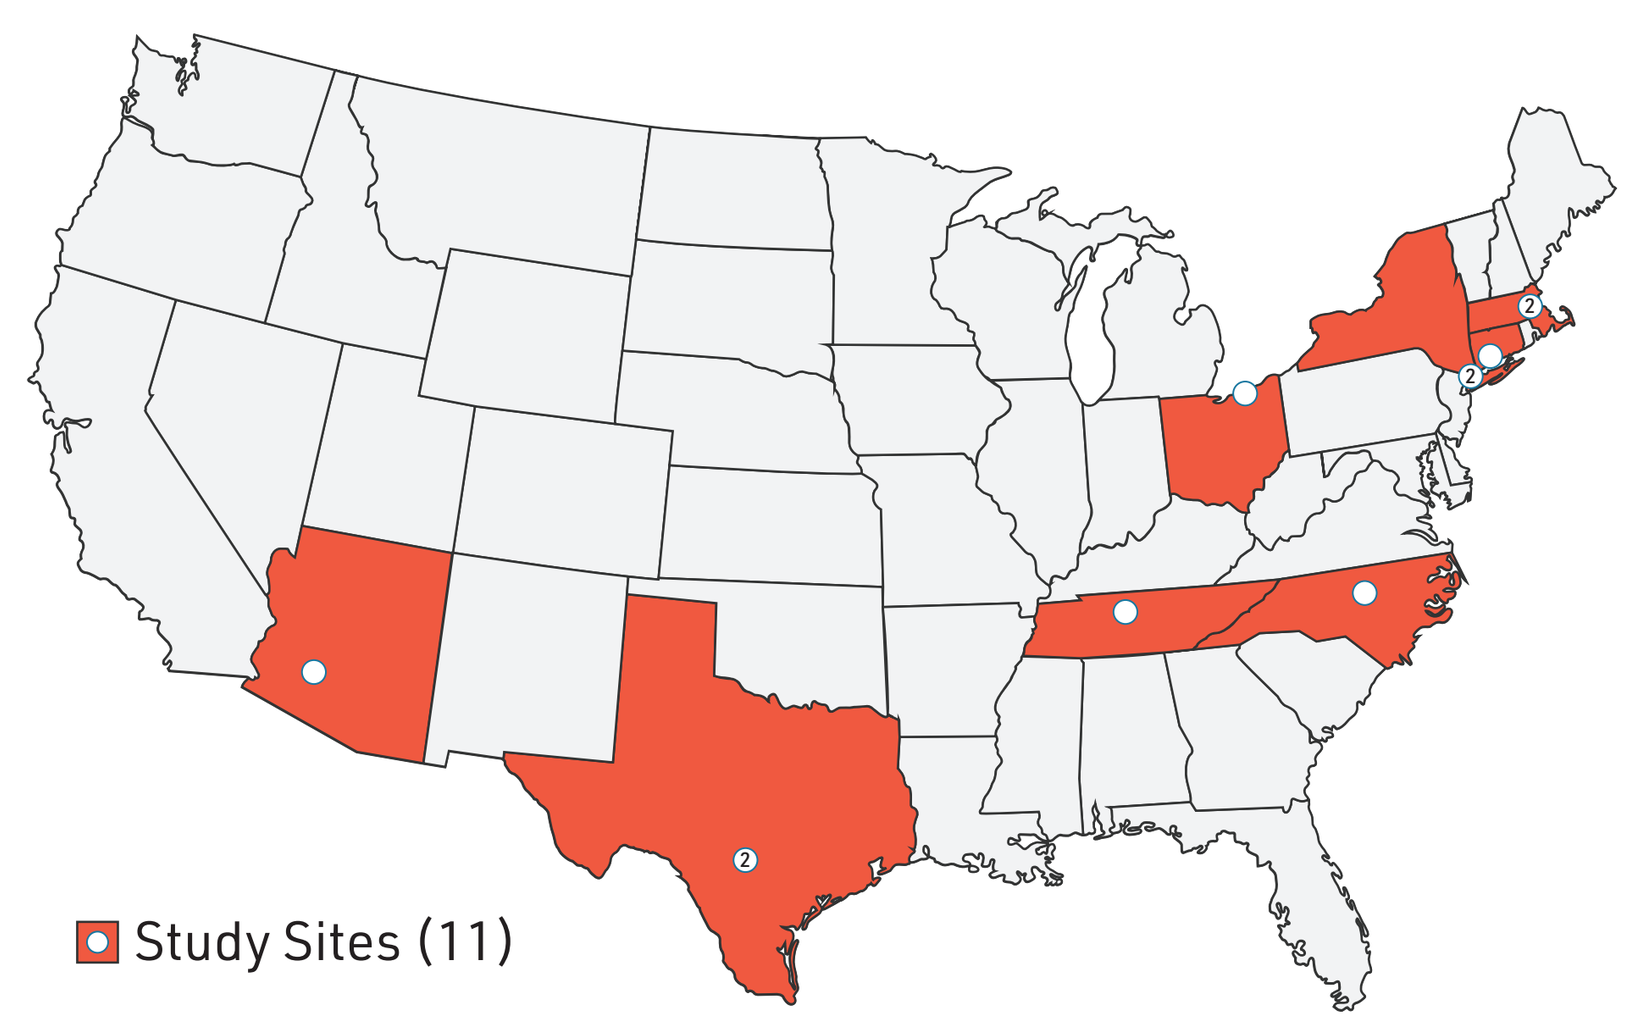
Study Sites (11)

PRECLINICAL EVALUATION OF FX-909

Key Properties

FX-909 is an irreversible covalent inverse agonist that specifically and potently inhibits PPARG.

Property	FX-909
Potency	Target gene suppression in cells IC_{50} = 1-2 nM
Selectivity	> 2,000-fold vs PPARA and PPARD
Absorption	Projected to be 70%
Metabolism	Hepatic; no unique human metabolites expected
Drug-Drug Interactions	No significant liability anticipated
Dosing	Projected clearance supports QD or BID dosing
Phase 1 Formulation	Tablets

FX-909 Drives Durable Tumor Regression in Preclinical Models at Very Low Oral Doses

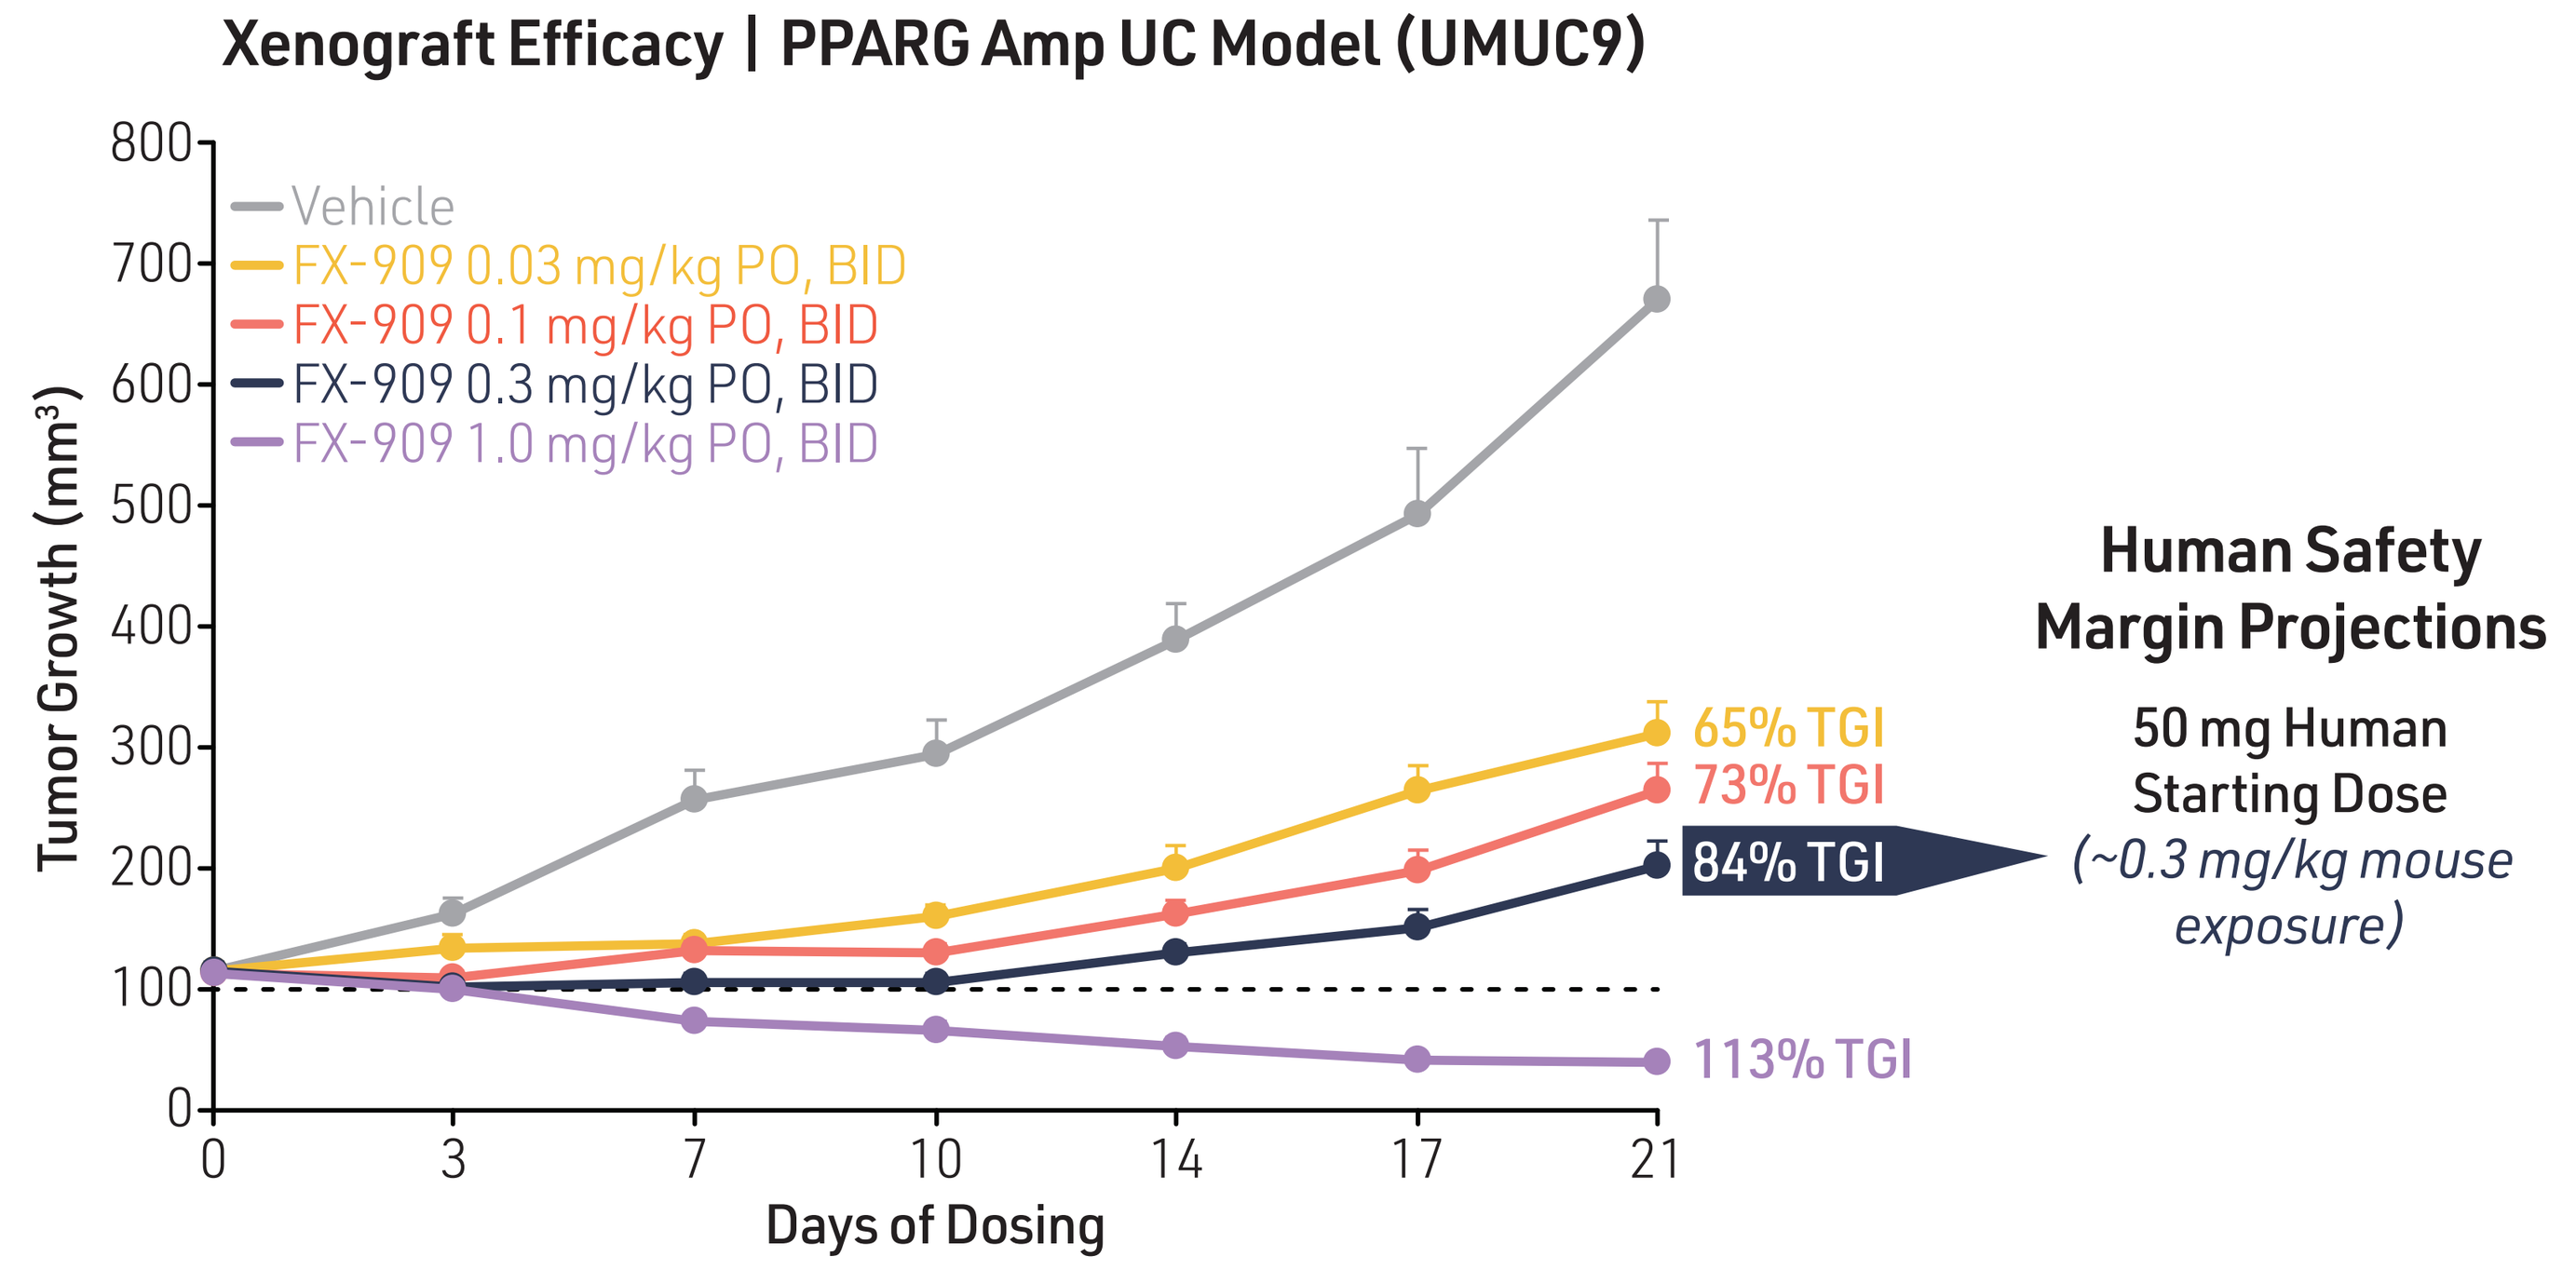
Xenograft Efficacy | PPARG Amp UC Model (UMUC9)
Vehicle
FX-909 0.03 mg/kg PO, BID
FX-909 0.1 mg/kg PO, BID
FX-909 0.3 mg/kg PO, BID
FX-909 1.0 mg/kg PO, BID
Tumor Growth (mm³)
Days of Dosing
0
100
200
300
400
500
600
700
800
3
7
10
14
17
21
65% TGI
73% TGI
84% TGI
113% TGI
Human Safety Margin Projections
50 mg Human Starting Dose
(~0.3 mg/kg mouse exposure)

Abbreviations: AE = adverse event; BID = twice daily; CNS = central nervous system; CrCl = creatinine clearance; ctDNA = circulating tumor DNA; CxDy = cycle x day y; DCR = disease control rate; DLT = dose-limiting toxicity; EOT = end of treatment; HbA1c = hemoglobin A1c; IC_{50} = half maximal inhibitory concentration; ORR = objective response rate; DOR = durability of response; OR = objective response; OS = overall survival; PD = pharmacodynamics; PFS = progression-free survival; PK = pharmacokinetics; PO = administered orally; QD = once daily; QTcF = corrected QT interval (via Fridericia formula); RP2D = recommended Phase 2 dose; SAE = significant adverse event; TGI = tumor growth inhibition; T1DM/T2DM = Type 1/2 diabetes mellitus.

References: 1) Surveillance, Epidemiology, and End Results Program. NCI. 2022. **2)** Powles T, et al. *N Engl J Med* 2020. **3)** Friedlander TW et al. 2021 ASCO Annual Meeting Abstract:4528. **4)** Varley CL et al. *J Cell Sci* 2004. **5)** Liu C et al. *Nat Commun* 2019. **6)** Biton A et al. *Cell Reports* 2014. **7)** Damrauer JS et al. *Proc Natl Acad Sci* 2014. **8)** Robertson AG et al. *Cell* 2017. **9)** Hedegaard J et al. *Cancer Cell* 2016. **10)** Motley W et al. 2022 EORTC-NCI-AACR, Abstract 333.

SUMMARY

- Activating alterations in PPARG/RXRA and FGFR3 are observed in 50% of patients with advanced luminal UC. Targeting of PPARG is a novel therapeutic target in advanced UC and offers hope for improving clinical outcomes in patients with a luminal subtype.
- FX-909-CLINPRO-1 (NCT05929235) is a first-in-human, multicenter, open-label Phase 1 study to evaluate the safety and tolerability, PK, PD, and preliminary clinical activity of the PPARG inverse agonist FX-909 given orally in patients with advanced solid malignancies.
- Exploratory objectives include the evaluation of patient selection biomarkers from tissue and blood samples and association with clinical outcomes.
- In the US, approximately 11 sites are planned for Phase 1A and 12-15 sites for Phase 1B.

ACKNOWLEDGMENTS

The authors thank the patients and their families as well as the investigators and participating study teams for making this study possible. All authors contributed and approved the presentation; professional medical writing and/or editorial assistance was provided by Paginae, Inc. This study is funded by Flare Therapeutics.

Scan to view this poster